The Oral P38a Kinase Inhibitor Neflamapimod Slows The Rate Of Clinical Worsening In Dementia With Lewy Bodies (DLB)

P1-001

John-Paul Taylor[1], John Alam[2], Stephen N. Gomperts[3], Lawrence Honig[4], Niels Prins[5], Hui-May Chu[6],Amanda Gardner[2], Kelly Blackburn[2], Charlotte Teunissen[7], James Galvin[8]

1. Translational and Clinical Research Institute and NIHR Newcastle Biomedical Research Centre, Newcastle University, Newcastle, United Kingdom, 2CervoMed, Inc., Boston, MA, USA, 3Department of Neurology, Massachusetts General Hospital, Boston, MA, USA,44. Taub Institute for Research on Alzheimer's Disease and the Aging Brain, Vagelos College of Physicians and Surgeons, New York,NY, USA, 5Brain Research Centre, Amsterdam, Netherlands, 6Anoixis Corporation, Natick, MA, USA, 7Neurochemistry Laboratory,Department of Laboratory Medicine, Amsterdam UMC, Vrije Universiteit Amsterdam, Amsterdam, Netherlands, 88. Comprehensive Center for Brain Health, Department of Neurology, University of Miami Miller School of Medicine, Boca Raton, FL, USA

In a 159-patient randomized trial in dementia with Lewy bodies, neflamapimod did not replicate phase 2a results in which it improved outcomes on CDR-SB versus placebo. In exploratory analyses of the current study, there was evidence that neflamapimod slowed worsening on CDR-SB in patients with low plasma p-tau181and higher plasma drug concentrations, key lessons ahead of advancing into phase 3 development.

INTRODUCTION

Phase 2b 16-Week Placebo-Controlled Clinical Trial, with 32-Week Neflamapimod Only Extension

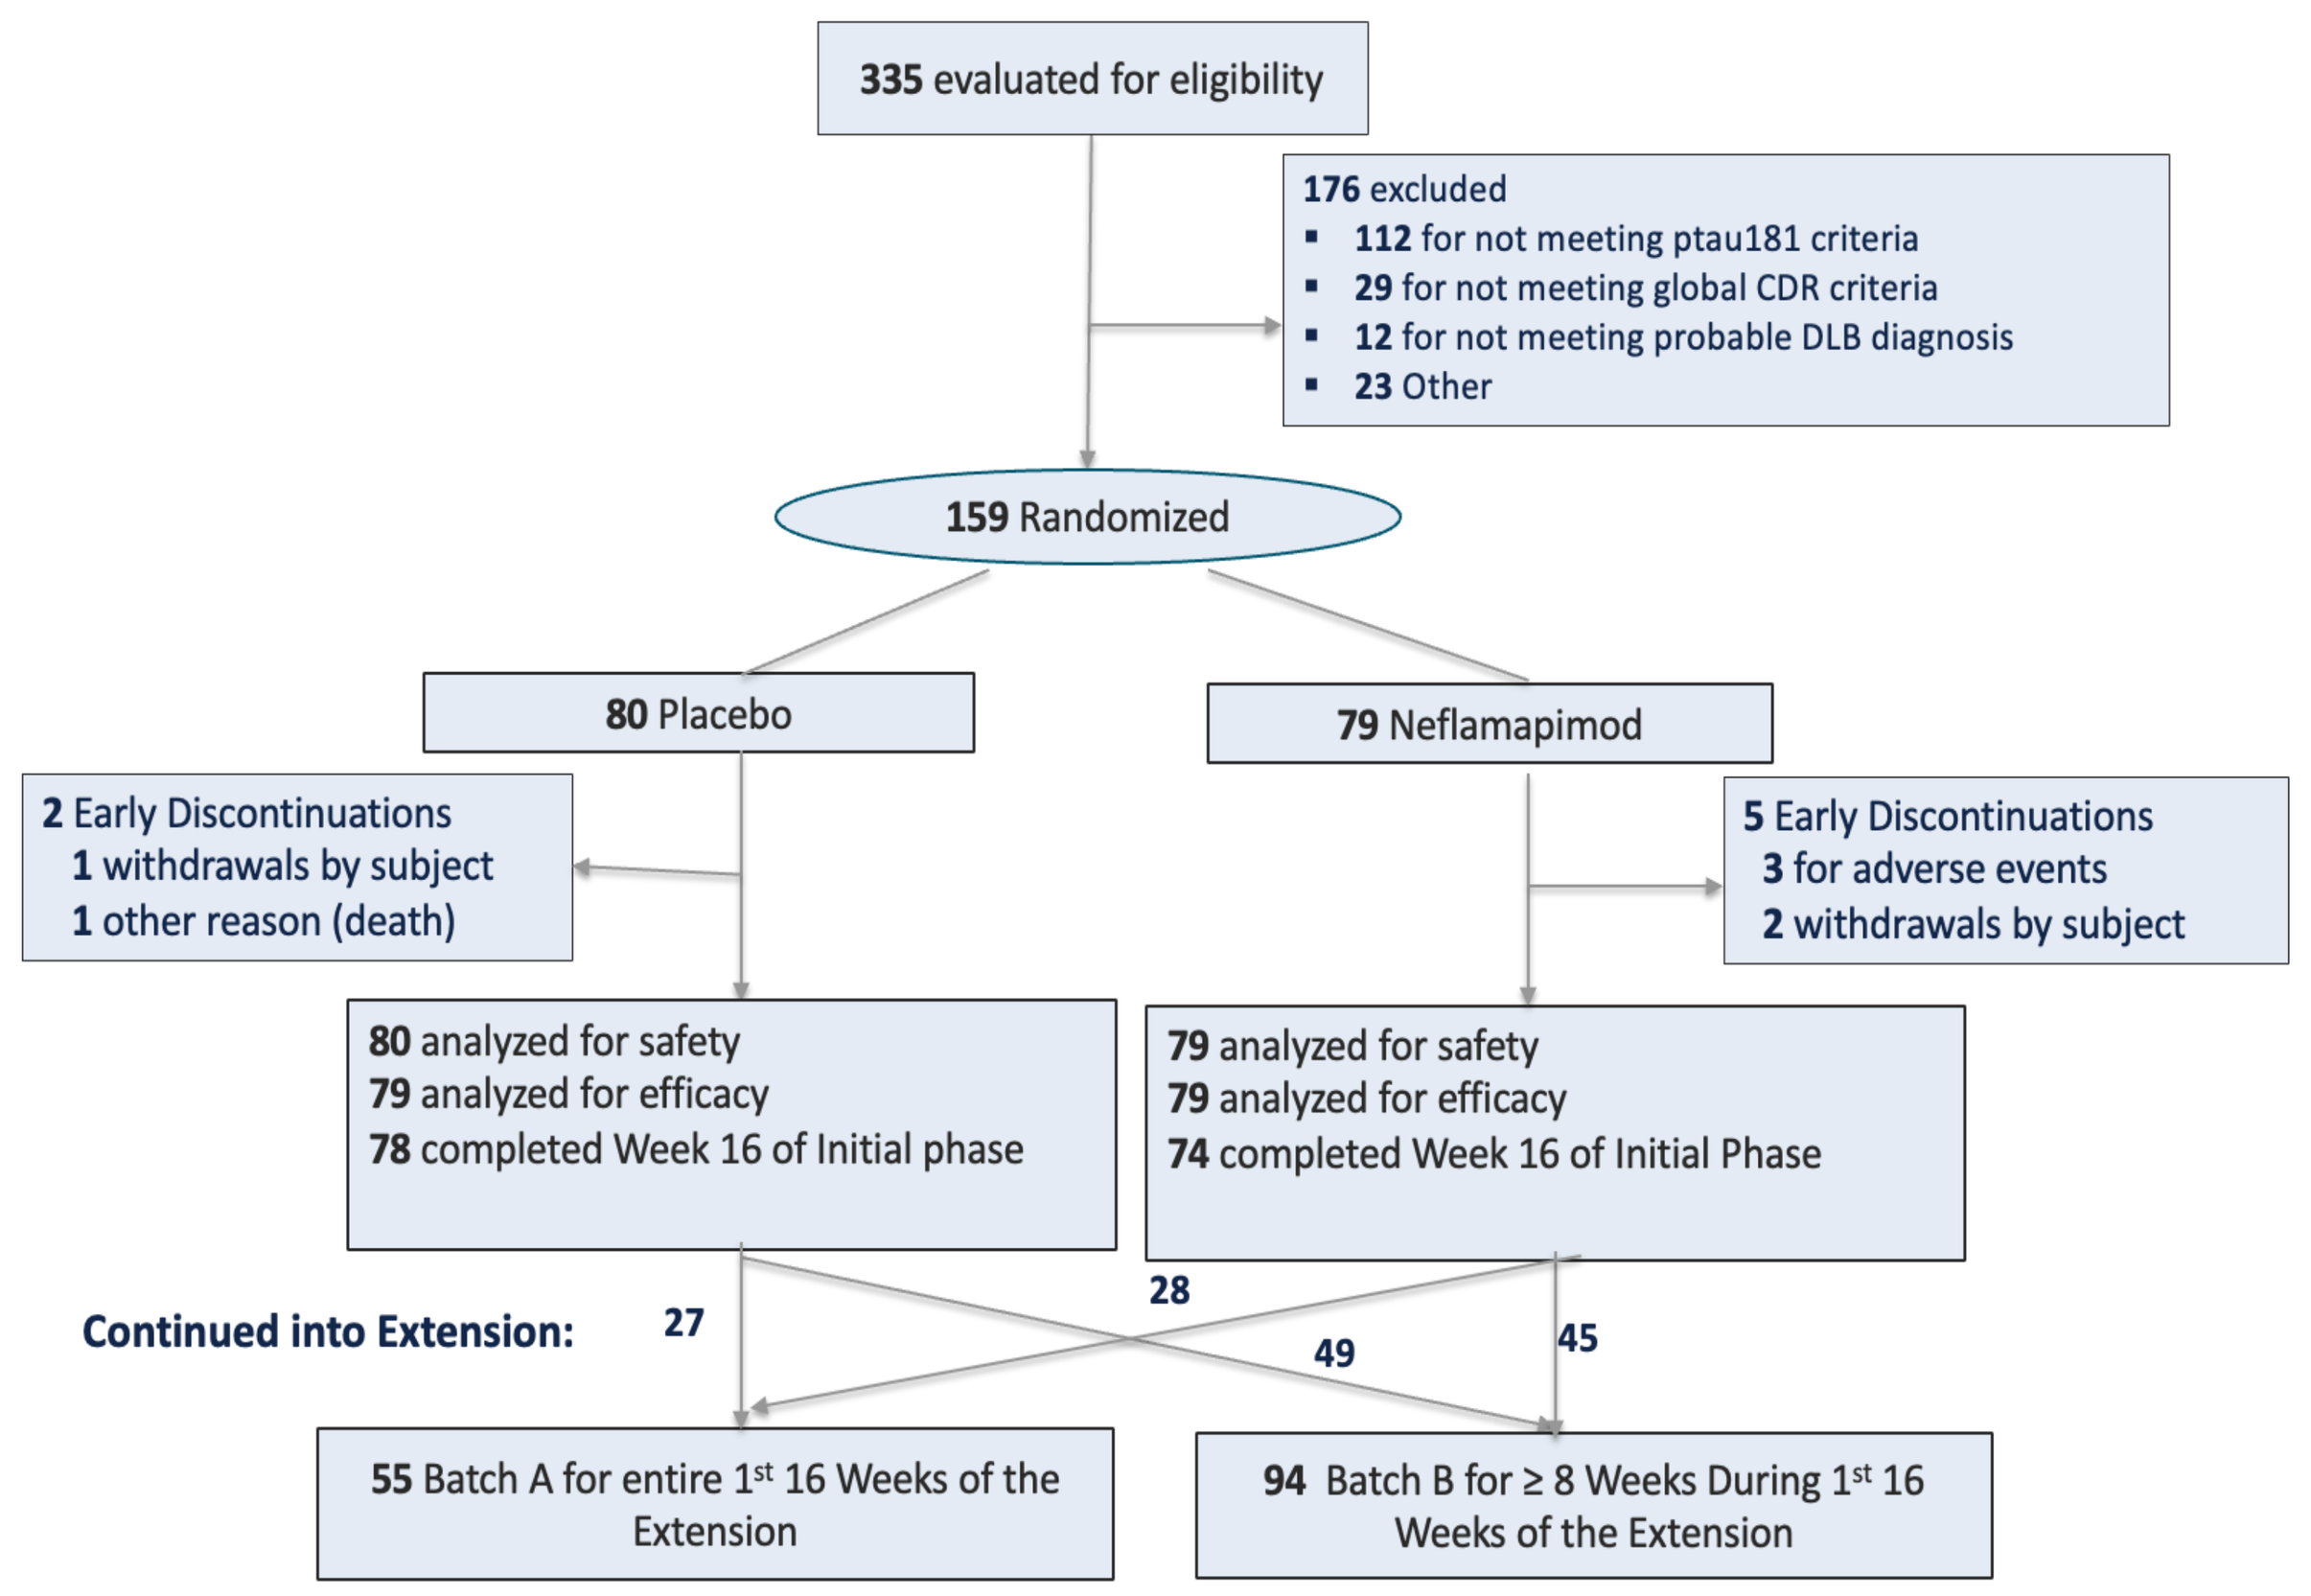
335 evaluated for eligibility
176 excluded
112 for not meeting ptau181 criteria
29 for not meeting global CDR criteria
12 for not meeting probable DLB diagnosis
23 Other
159 Randomized
80 Placebo
79 Neflamapimod
2 Early Discontinuations
1 withdrawals by subject
1 other reason (death)
5 Early Discontinuations
3 for adverse events
2 withdrawals by subject
80 analyzed for safety
79 analyzed for efficacy
78 completed Week 16 of Initial phase
79 analyzed for safety
79 analyzed for efficacy
74 completed Week 16 of Initial Phase
Continued into Extension:
27
28
49
45
55 Batch A for entire 1st 16 Weeks of the Extension
94 Batch B for ≥ 8 Weeks During 1st 16 Weeks of the Extension

Baseline Characteristics

- All participants met consensus clinical criteria for probable DLB

	Placebo	Neflamapimod
Number of Participants	79	80
Age (M, SD)	70.7 (5.8)	72.1 (6.4)
Male	67 (83.8%)	69 (87.3%)
MMSE (M, SD)	23.3 (4.6)	23.6 (4.3)
CDR-SB (M, SD)	4.23 (1.71)	4.45 (2.17)
ISLT Immediate (M, SD)	14.8 (5.9)	13.0 (5.0)
Core Clinical Criteria:		
Cognitive fluctuations	59 (73.8%)	59 (74.7%)
Visual Hallucinations	45 (56.3%)	43 (54.4%)
REM sleep behavioral disorder	64 (80.0%)	60 (75.9%)
Parkinsonism	68 (85.0%)	70 (88.6%)
Background Therapy		
AChEI alone*	52 (65.0%)	50 (63.3%)
Memantine (with or without AChEI)	11 (13.8%)	12 (15.2%)
No background therapy	17 (21.3%)	17 (21.5%)
Screening plasma ptau181 level (pg/mL)	19.1 (5.2)	18.9 (4.9)
Number (%) with screening plasma ptau181<21 pg/mL	46 (61%)	48 (60%)

RESULTS

Primary Endpoint Results

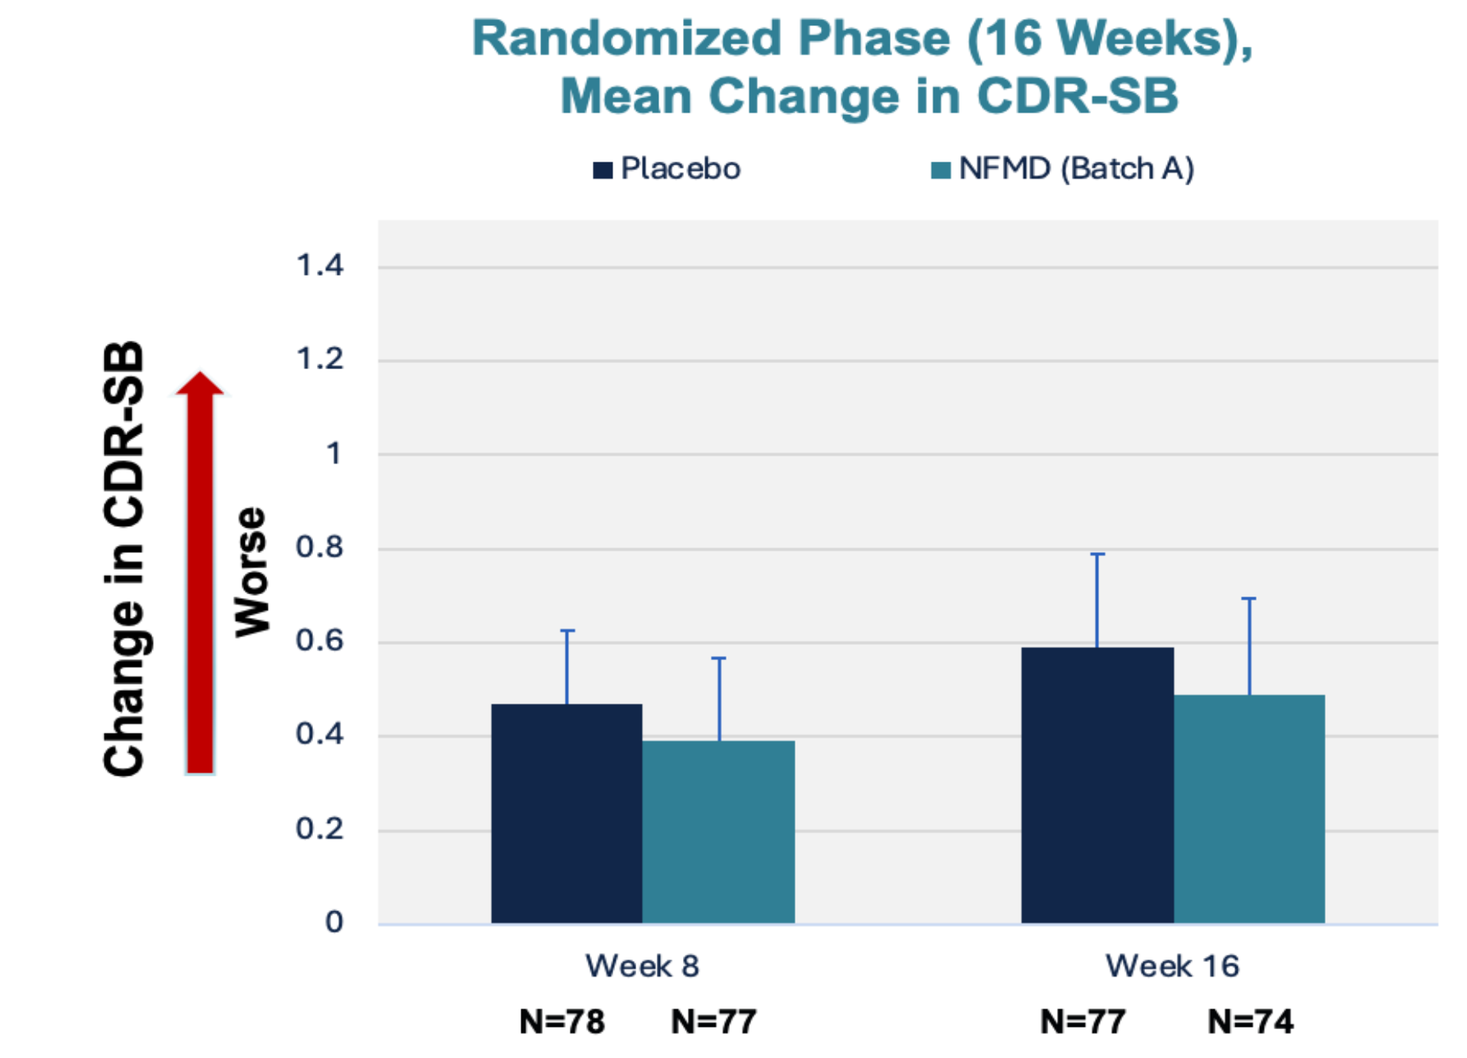
Randomized Phase (16 Weeks), Mean Change in CDR-SB
Placebo
NFMD (Batch A)
Change in CDR-SB
Worse
1.4
1.2
1
0.8
0.6
0.4
0.2
0
Week 8
Week 16
N=78 N=77
N=77 N=74

The Phase 2a clinical study findings were not replicated. However, **a higher proportion of patients with AD co-pathology** than targeted were included and neflamapimod **drug product did not achieve expected plasma drug concentrations.**

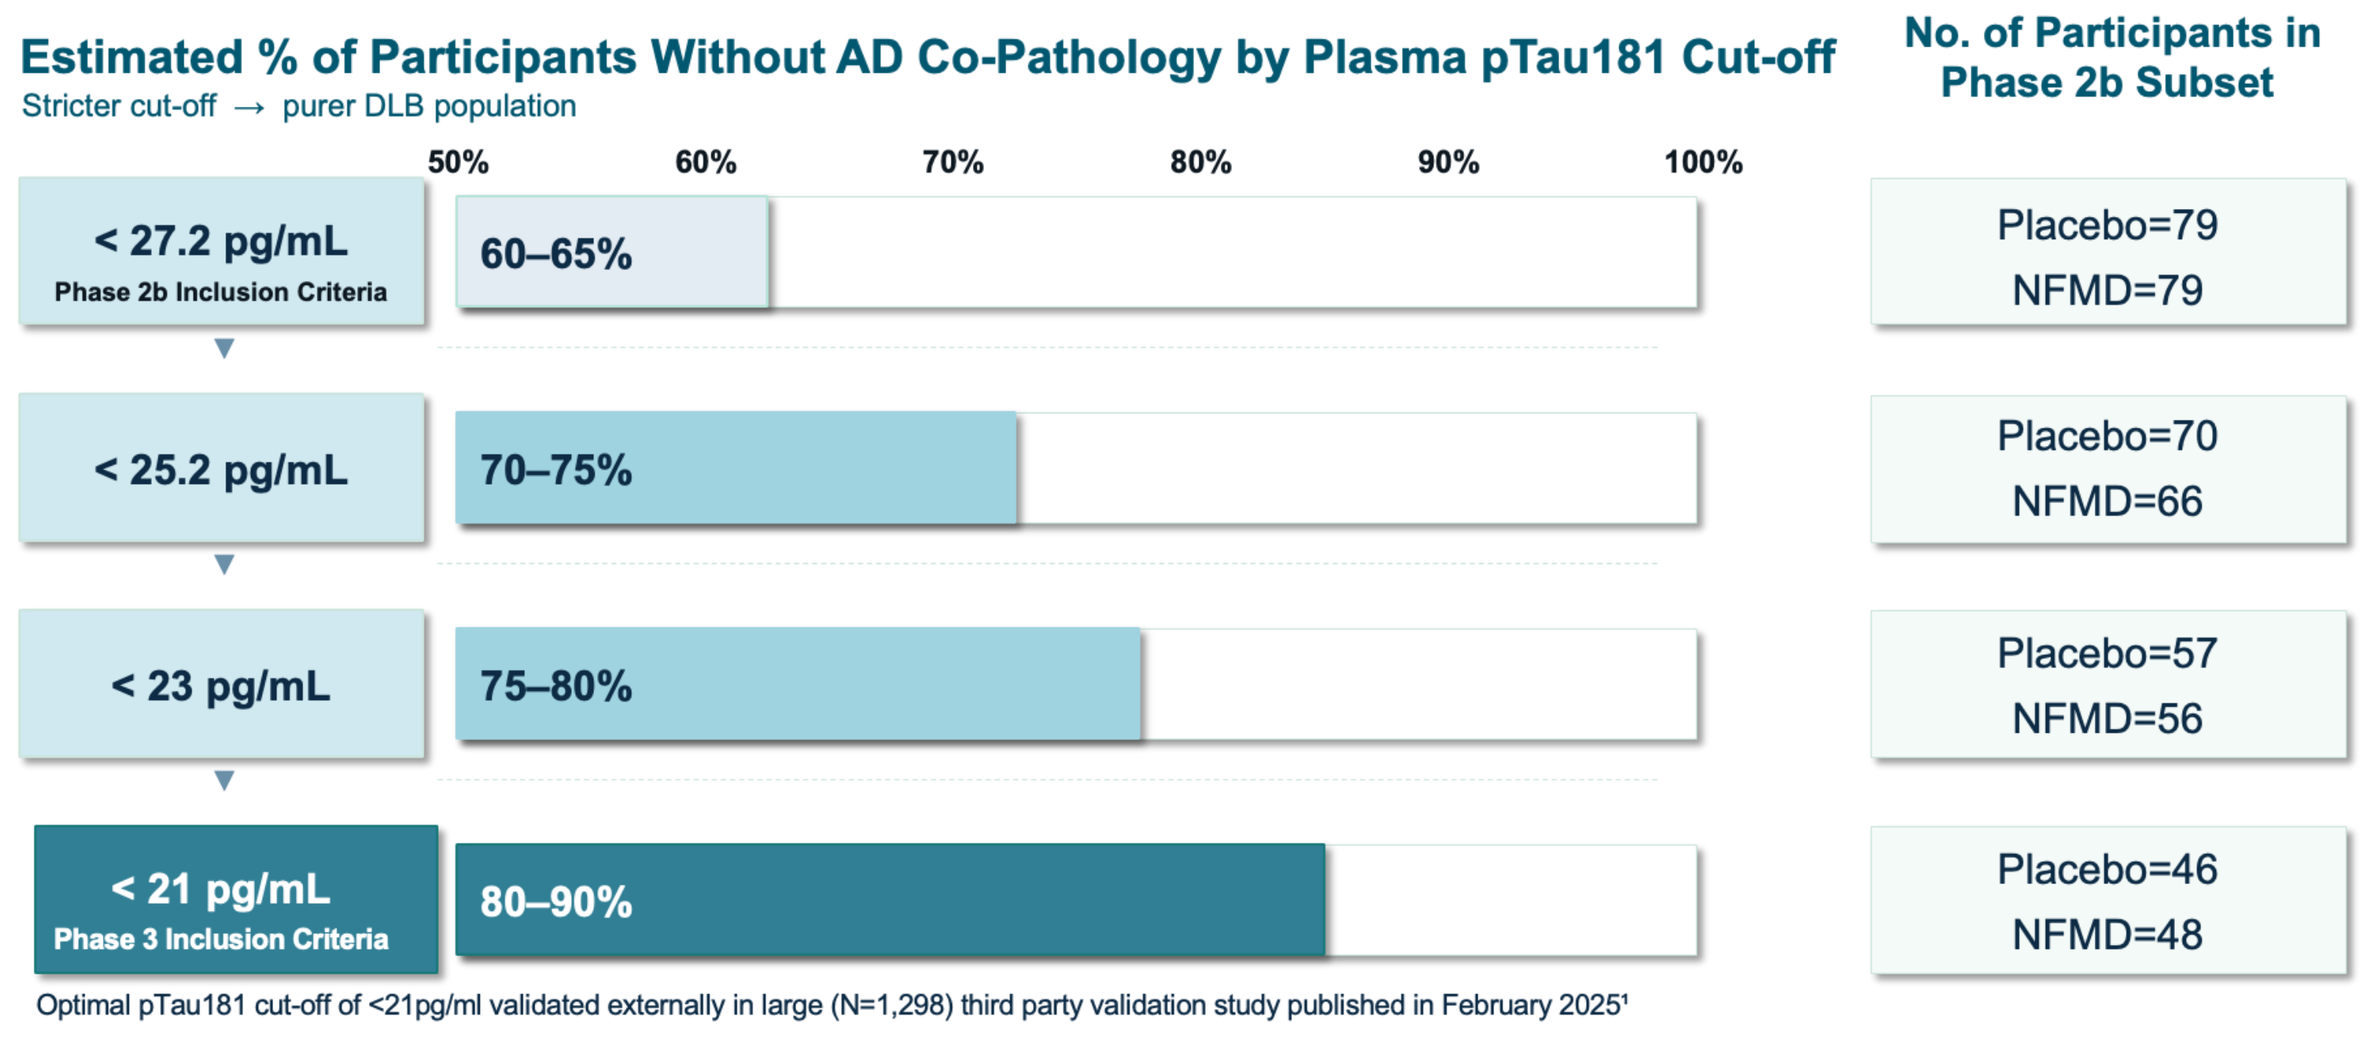
Estimated % of Participants Without AD Co-Pathology by Plasma pTau181 Cut-off
Stricter cut-off → purer DLB population
50%
60%
70%
80%
90%
100%
< 27.2 pg/mL
Phase 2b Inclusion Criteria
60–65%
< 25.2 pg/mL
70–75%
< 23 pg/mL
75–80%
< 21 pg/mL
Phase 3 Inclusion Criteria
80–90%
No. of Participants in Phase 2b Subset
Placebo=79
NFMD=79
Placebo=70
NFMD=66
Placebo=57
NFMD=56
Placebo=46
NFMD=48
Optimal pTau181 cut-off of <21pg/ml validated externally in large (N=1,298) third party validation study published in February 2025¹

Neflamapimod Effect vs. Placebo on Change in CDR-SB by Plasma pTau181 Cutoff (MMRM Analysis)

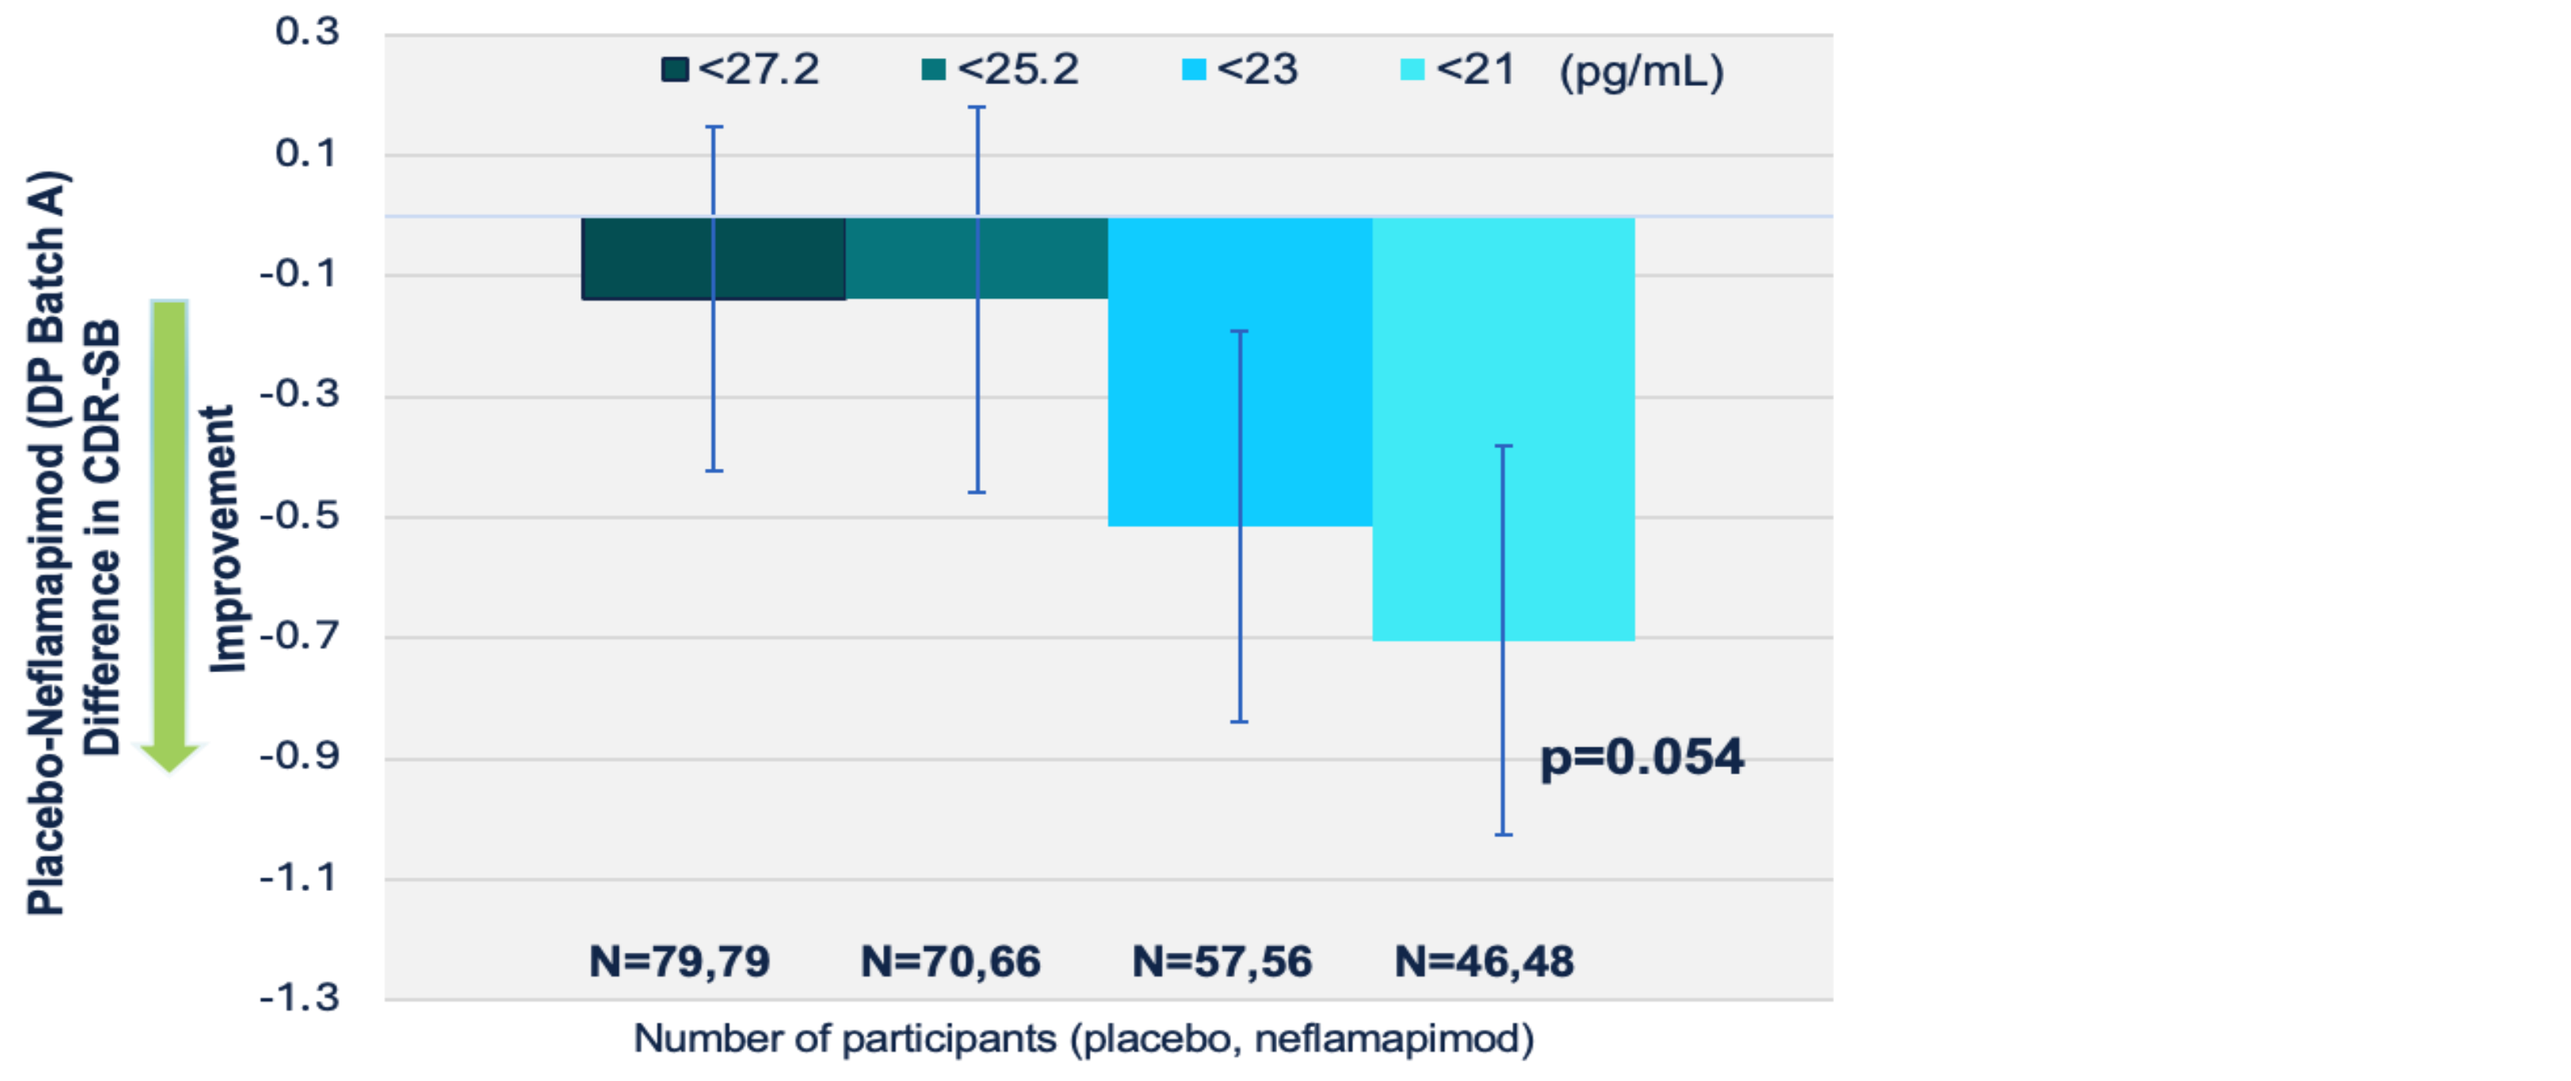
<27.2
<25.2
<23
<21 (pg/mL)
Placebo-Neflamapimod (DP Batch A) Difference in CDR-SB
Improvement
0.3
0.1
-0.1
-0.3
-0.5
-0.7
-0.9
-1.1
-1.3
p=0.054
N=79,79
N=70,66
N=57,56
N=46,48
Number of participants (placebo, neflamapimod)

Change in CDR-SB During Placebo Controlled Phase by Trough Plasma Drug Concentration (C_{trough}) in <21 pTau181 Subset

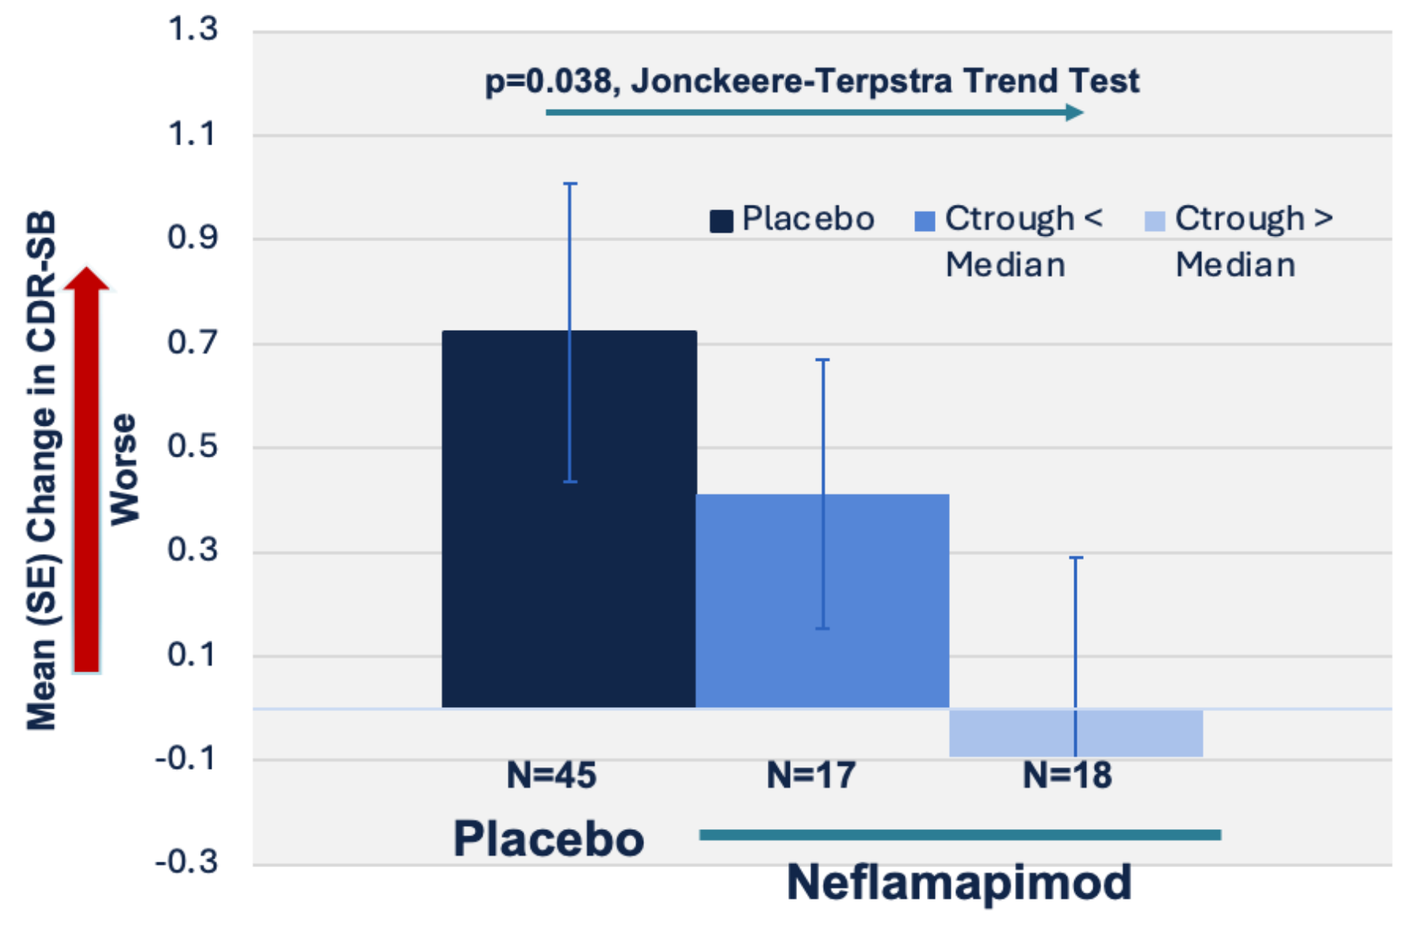
p=0.038, Jonckeere-Terpstra Trend Test
Placebo
Ctrough < Median
Ctrough > Median
Mean (SE) Change in CDR-SB
Worse
1.3
1.1
0.9
0.7
0.5
0.3
0.1
-0.1
-0.3
N=45
N=17
N=18
Placebo
Neflamapimod

Median Observed C_{trough} = 3.64 pg/mL

Neflamapimod Plasma Drug Concentration Were Lower Than Expected with Batch A During Randomized Phase and At Expected Levels with Batch B During the Extension Phase

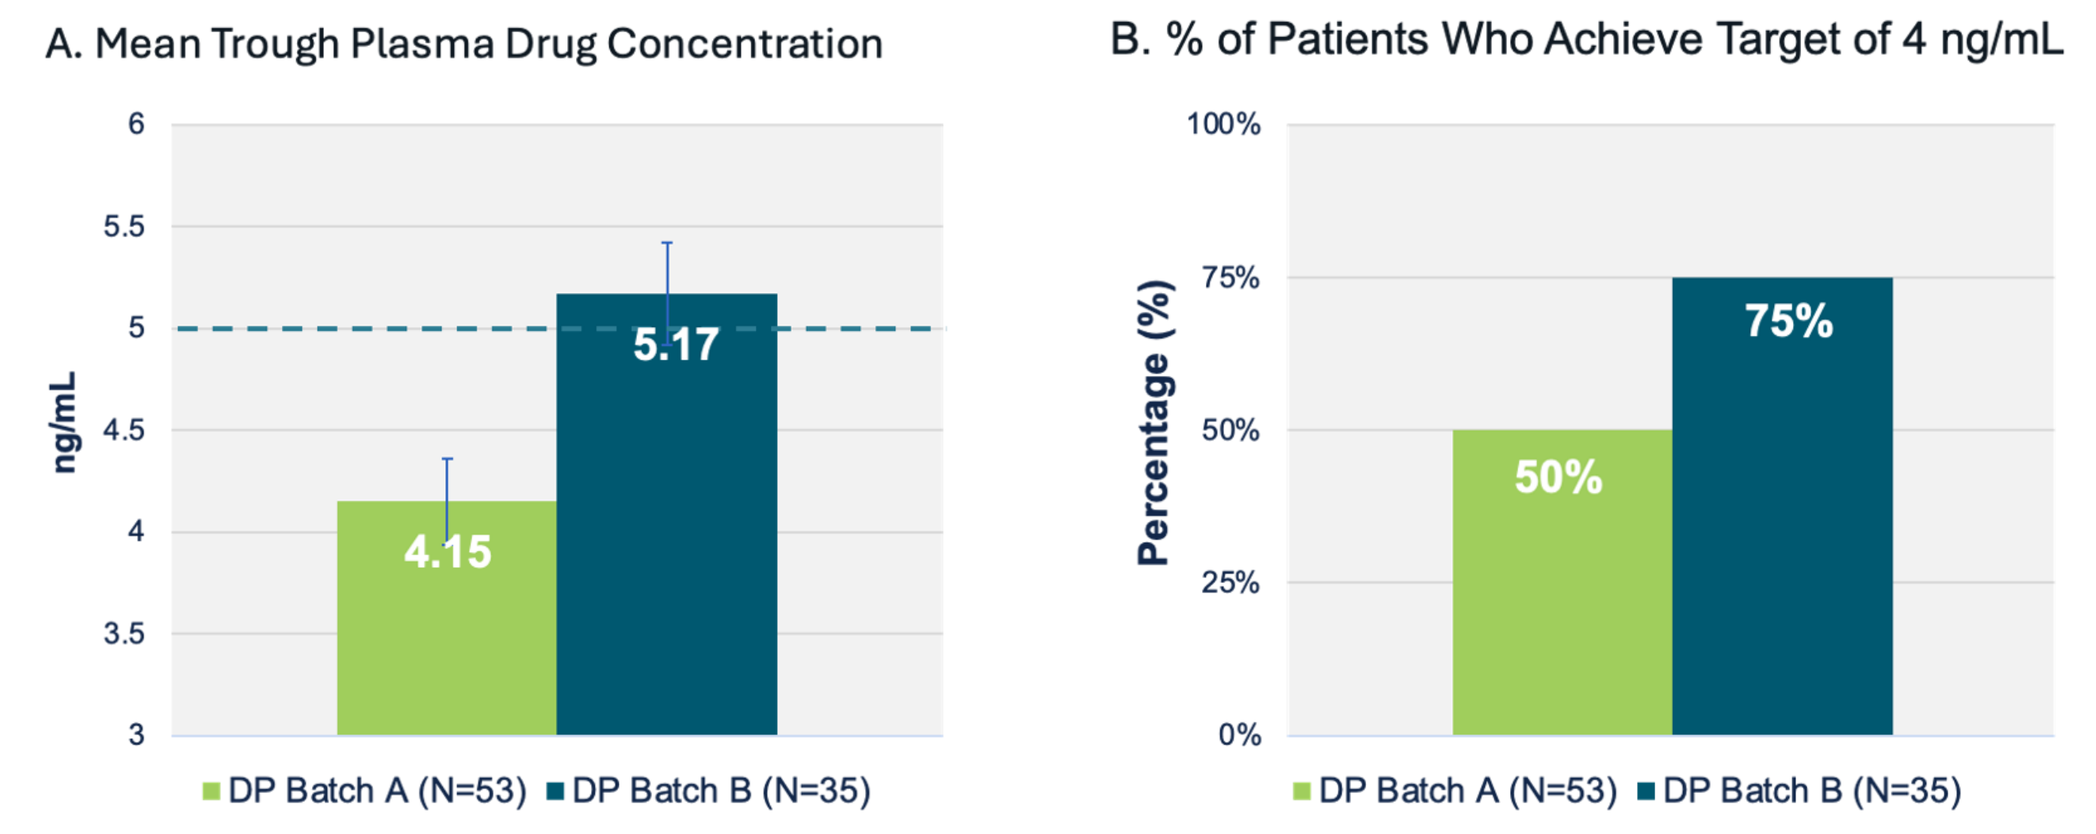
A. Mean Trough Plasma Drug Concentration
ng/mL
6
5.5
5
4.5
4
3.5
3
4.15
5.17
DP Batch A (N=53)
DP Batch B (N=35)
B. % of Patients Who Achieve Target of 4 ng/mL
Percentage (%)
100%
75%
50%
25%
0%
50%
75%
DP Batch A (N=53)
DP Batch B (N=35)

With Achieving Expected Plasma Drug Concentrations, Batch B Demonstrates Improvement on Change in CDR-SB in Within-Participant Comparison to Placebo

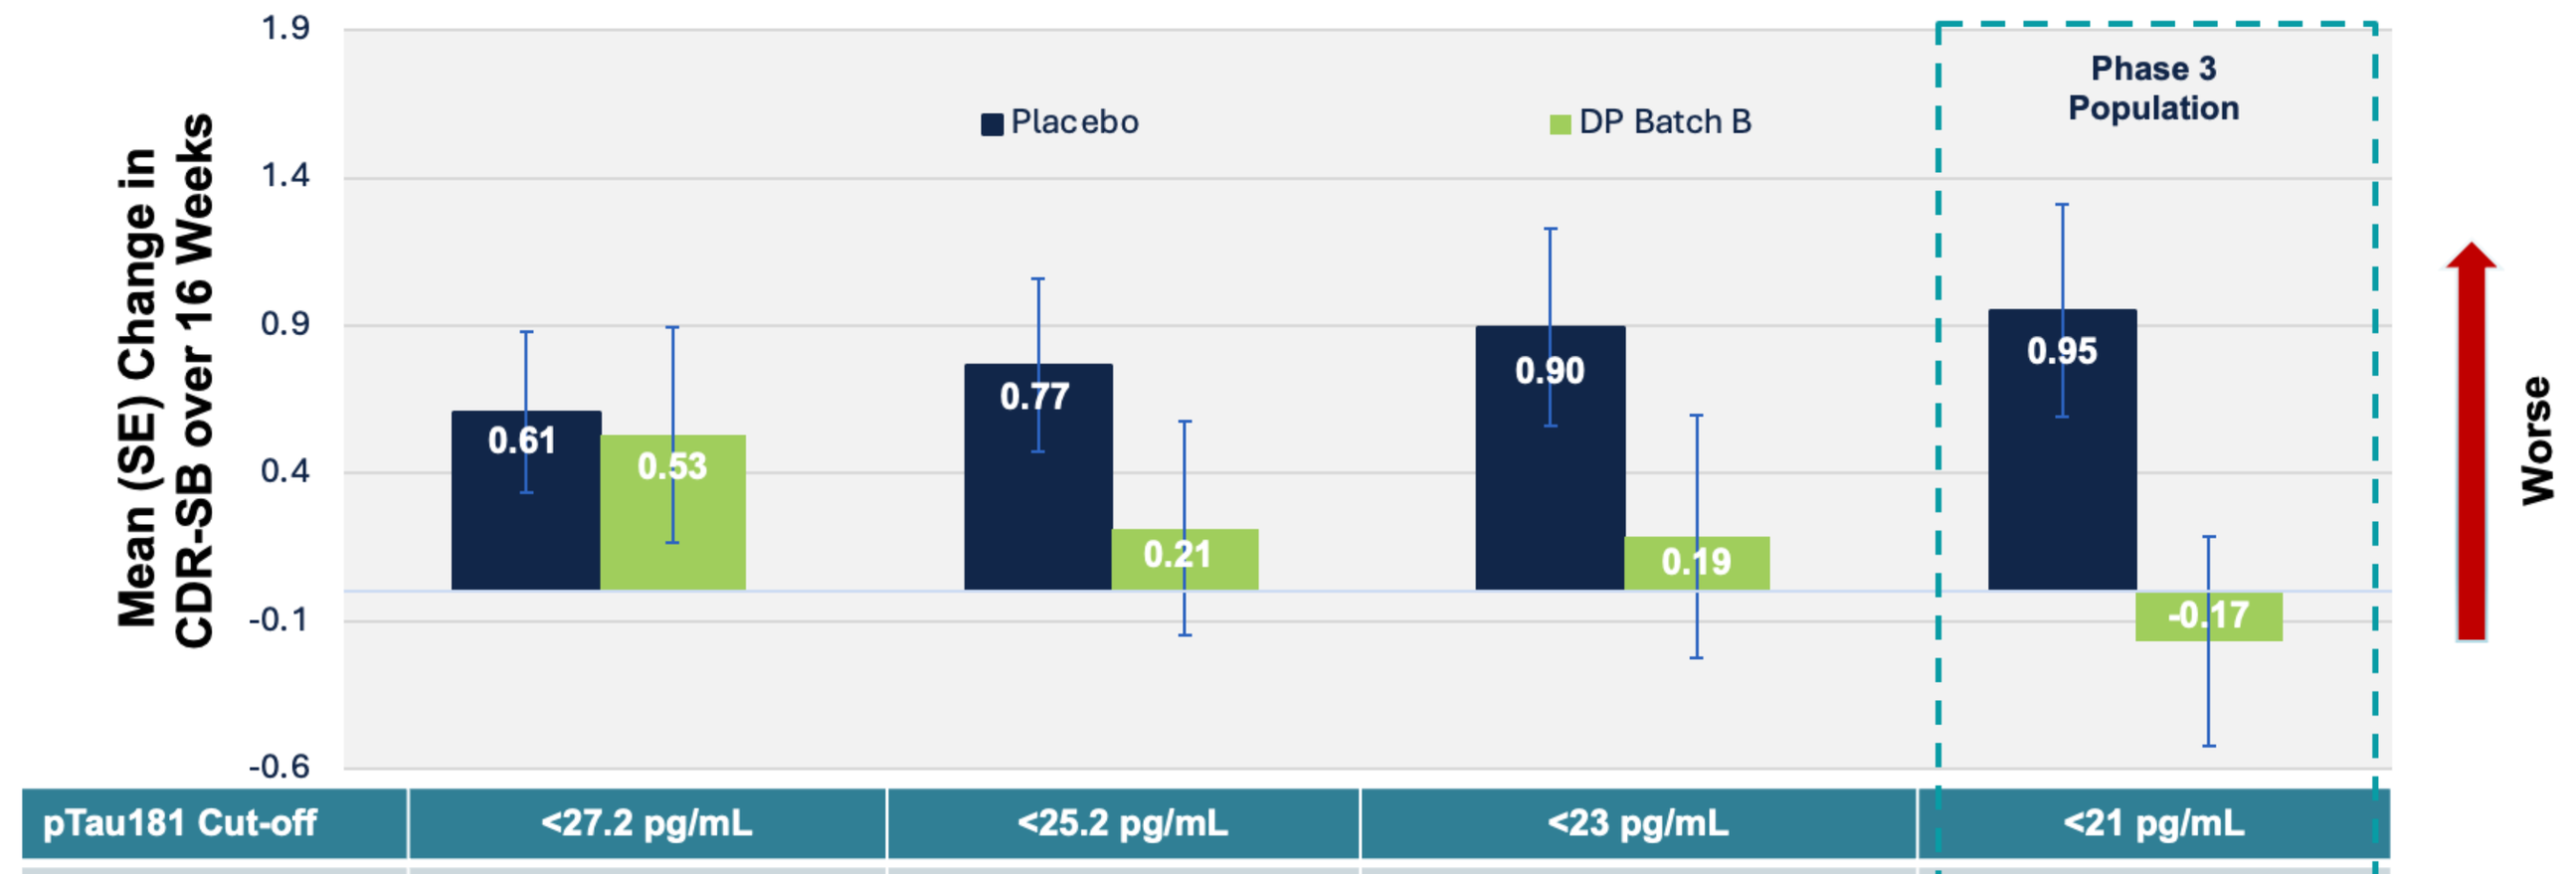
Mean (SE) Change in CDR-SB over 16 Weeks
Placebo
DP Batch B
Phase 3 Population
Worse
1.9
1.4
0.9
0.4
-0.1
-0.6
0.61
0.53
0.77
0.21
0.90
0.19
0.95
-0.17

pTau181 Cut-off	<27.2 pg/mL	<25.2 pg/mL	<23 pg/mL	<21 pg/mL
Number of Participants	32	28	24	21
NFMD-Placebo Difference	**-0.08**	**-0.55**	**-0.71**	**-1.11**
P-value NFMD vs. Placebo	p=0.9	p=0.044	p=0.034	p=0.005

CONCLUSIONS

- The Phase 2a study demonstrated improvement versus placebo on CDR-SB, particularly in participants with "pure" DLB (without AD co-pathology); this effect was not replicated in the current study.
- Exploratory analyses identified treatment effects favoring neflamapimod on CDR-SB during the placebo-controlled period among participants with low screening plasma p-tau181 (<21 pg/mL) and among those with trough plasma concentrations (C_{trough}) above the study median. These findings suggest that the negative primary outcome was driven by inclusion of a higher-than-targeted proportion of participants with AD co-pathology and lower-than-expected neflamapimod exposure.
- The optimal Phase 3 population is DLB without AD co-pathology, defined by plasma p-tau181 <21 pg/mL (v2.1 Simoa assay), a threshold recently identified as the high- sensitive cutoff for AD pathology (Doecke et al., 2025).
- The findings with Batch B further support that lower-than-expected neflamapimod exposure contributed to the primary result, as well support the choice of 50mg TID as the dose for phase 3.

ALZHEIMER'S® ASSOCIATION | AAIC 26