The P38alpha Kinase Inhibitor Neflamapimod Has A Beneficial Effect On Basal Forebrain Atrophy Assessed By MRI In Dementia With Lewy Bodies

Monday
#1231

John Alam1,1, Ismail Koubiyr2,3, Stephen N. Gomperts4,5, Menno M. Schoonheim4,6.
1CervoMed, Inc., Boston, MA, USA, 2Amsterdam UMC, Vrije Universiteit, Amsterdam, Netherlands, 3Amsterdam Medical Center, Amsterdam, Netherlands, 45Massachusetts General Hospital, Boston, MA, USA, 6Department of Anatomy and Neurosciences, Amsterdam Medical Center, Amsterdam, Netherlands.

Neflamapimod treatment increased right basal forebrain (BF) volume relative to placebo over the 16 weeks of the randomized period of the study, an effect that was maintained during an additional 32 weeks of open label treatment. In addition, during the open label extension, 32 weeks treatment increased functional connectivity between right BF and the right dorsal mode network (DMN)

INTRODUCTION

Background: Neflamapimod, an oral p38α inhibitor, targets molecular mechanisms underlying cholinergic degeneration and in preclinical studies reversed the cholinergic degenerative process in the basal forebrain (BF) and normalized performance in behavioral tests associated with cholinergic function. In phase 2a and 2b ("RewinD-LB") trials, neflamapimod slowed clinical progression in dementia with Lewy bodies (DLB), a disorder characterized by loss of basal forebrain cholinergic neurons. BF atrophy by MRI is correlated to cognitive decline in DLB and in an uncontrolled exploratory study in early AD (Prins et al, 2024)neflamapimod increased the volume of the Nucleus basalis of Meynert (NbM) and its functional connectivity, as assessed by structural and functional MRI, respectively. Herein we report on the MRI results from RewinD-LB.

Design

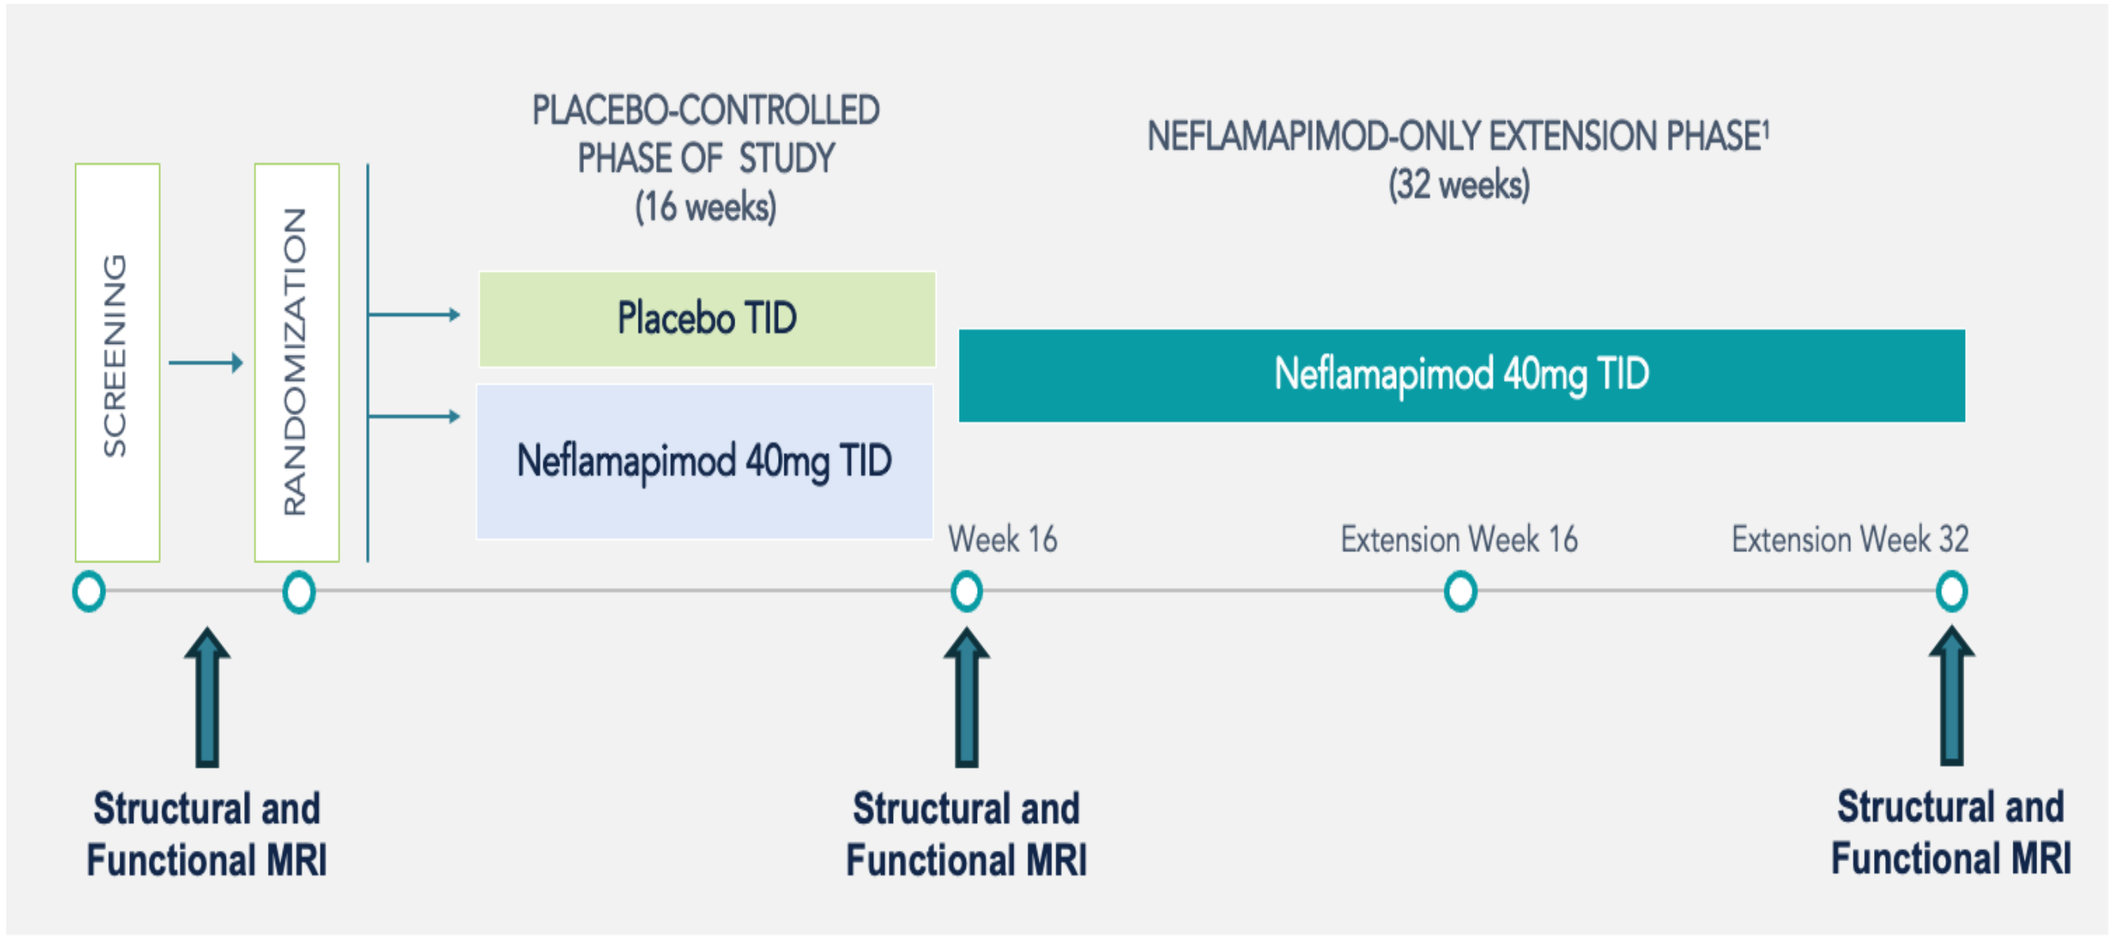
SCREENING
RANDOMIZATION
PLACEBO-CONTROLLED PHASE OF STUDY (16 weeks)
NEFLAMAPIMOD-ONLY EXTENSION PHASE¹ (32 weeks)
Placebo TID
Neflamapimod 40mg TID
Neflamapimod 40mg TID
Week 16
Extension Week 16
Extension Week 32
Structural and Functional MRI
Structural and Functional MRI
Structural and Functional MRI

Structural and functional MRI were performed at baseline, week 16, and week 48 in the 25participants from the UK and the Netherlands. N=10 placebo, 8 Neflamapimod during placebo-controlled phase; N=11 during the extension

RESULTS

Change from Baseline in NbM or BF Volume Baseline During the 16-Week Duration Placebo-Controlled Phase

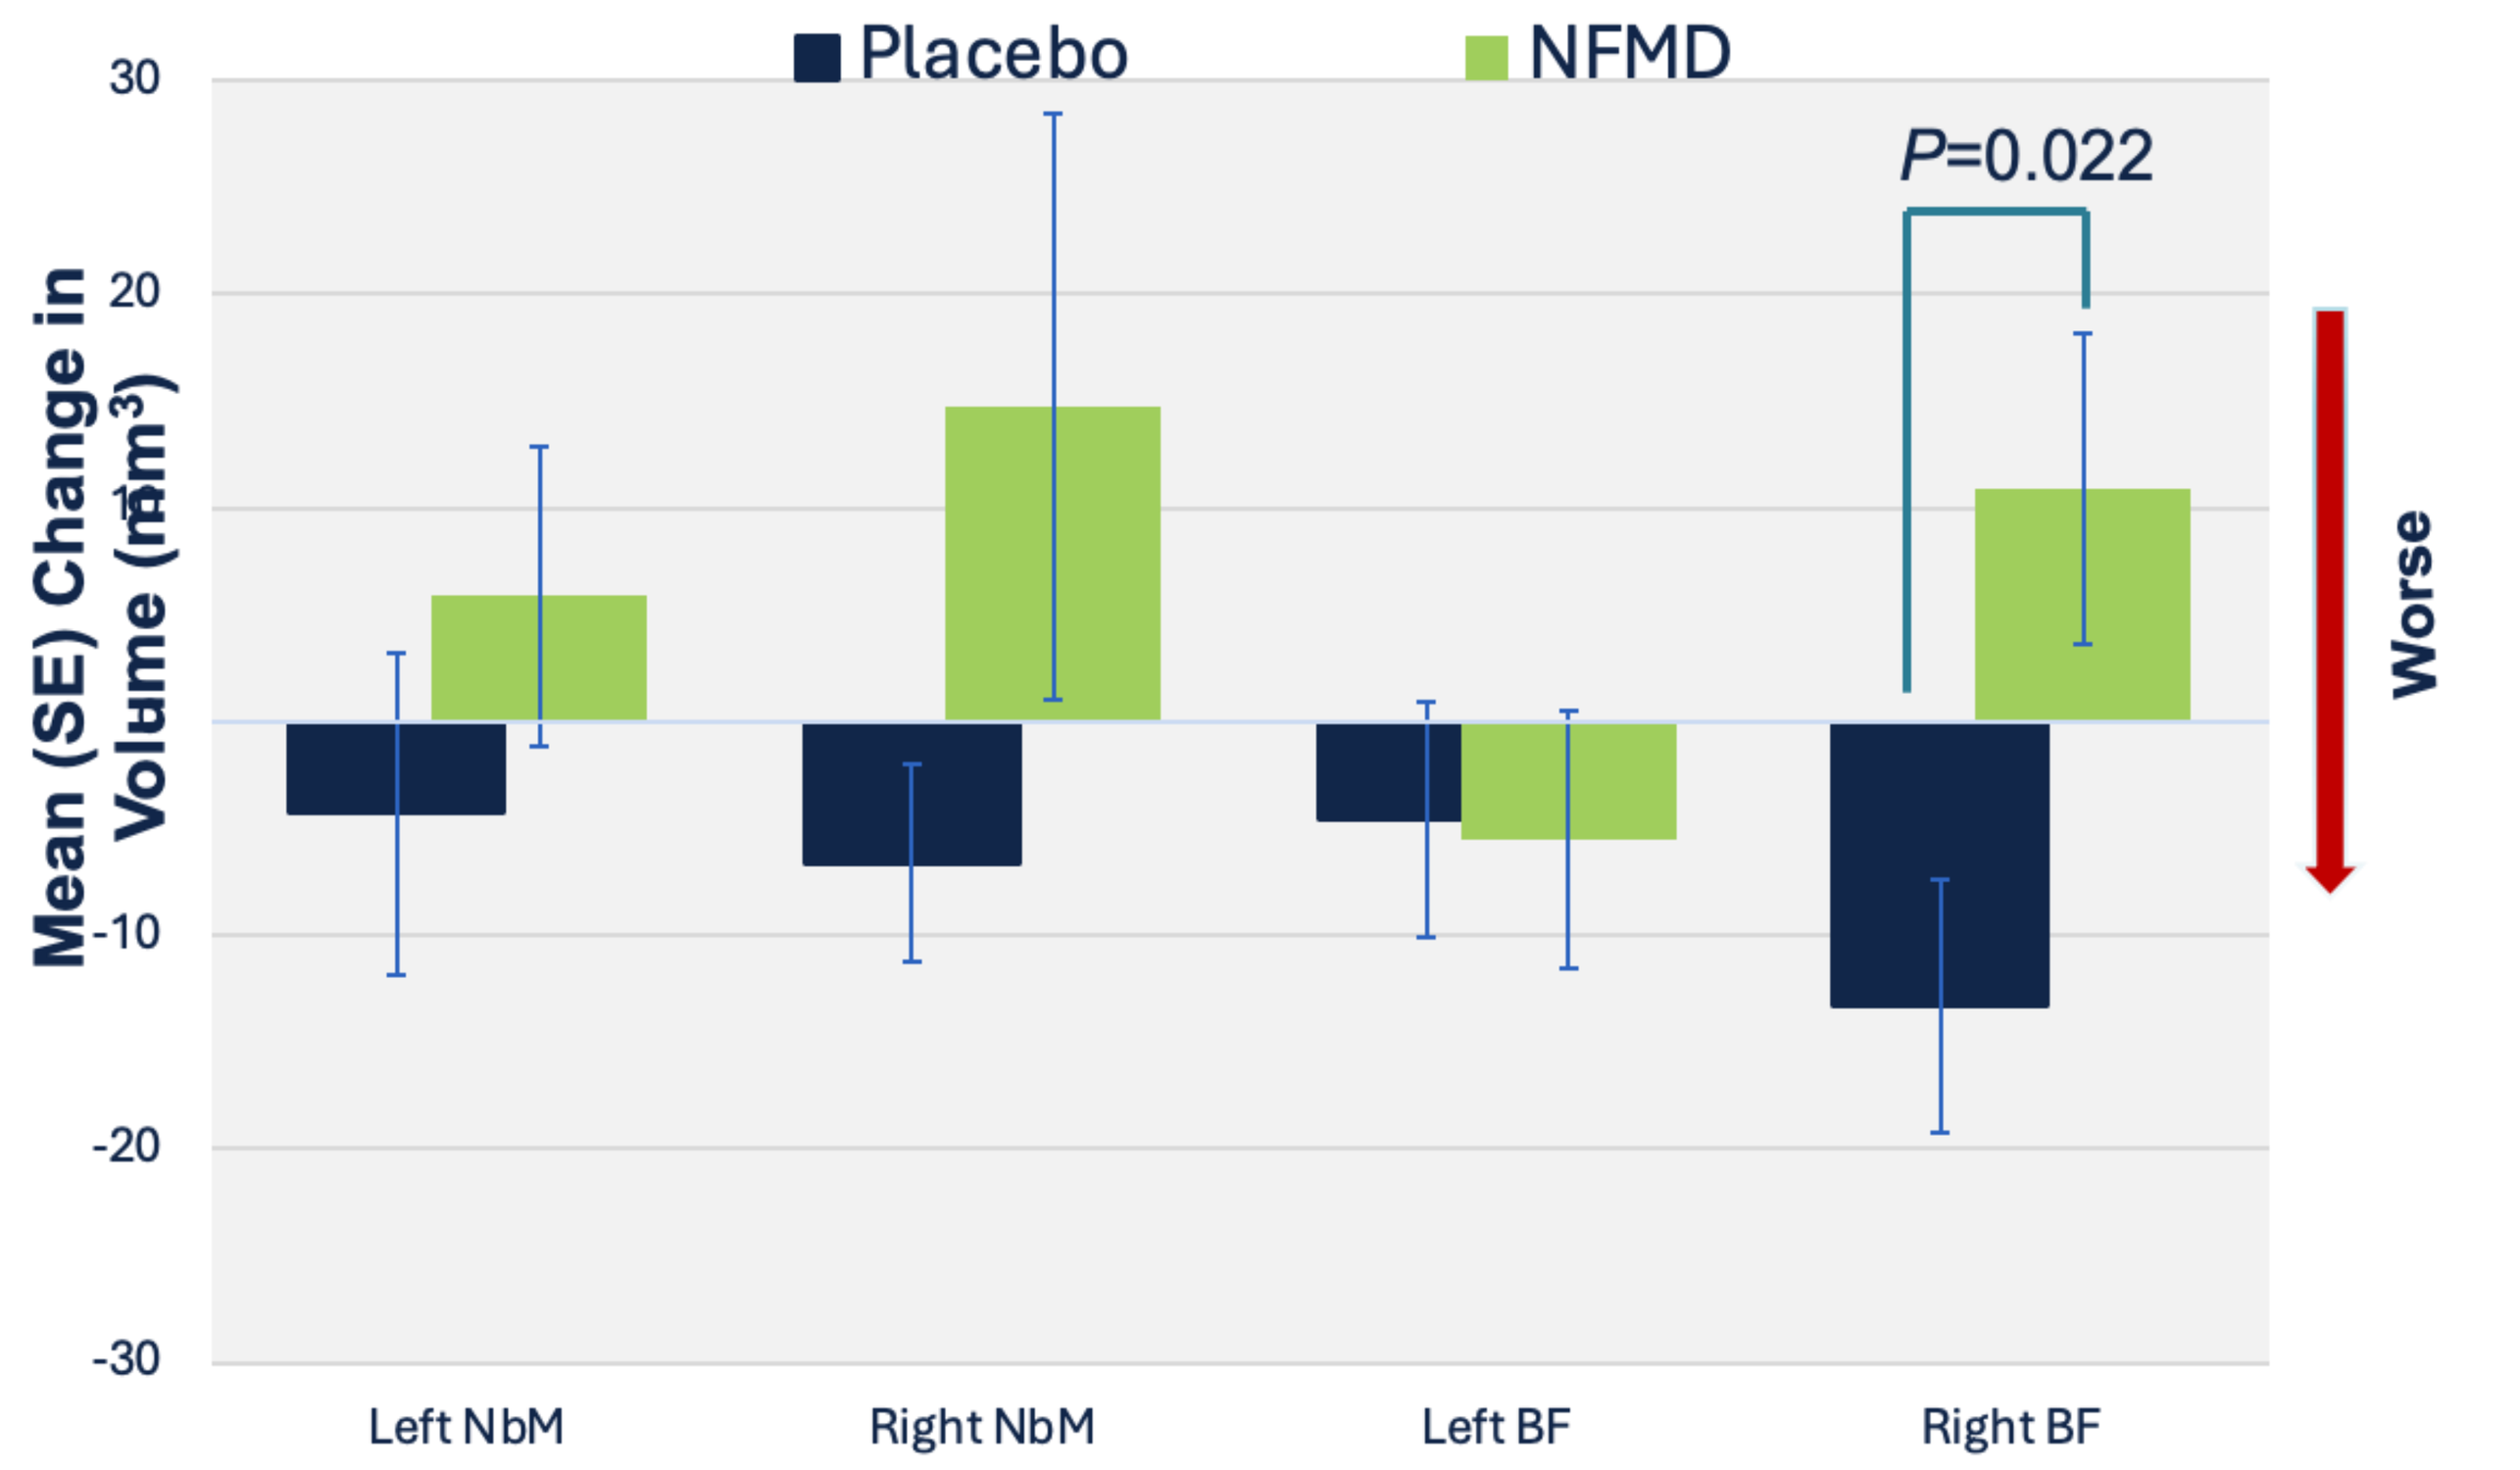
Placebo
NFMD
P=0.022
Mean (SE) Change in Volume (mm³)
30
20
10
0
-10
-20
-30
Left NbM
Right NbM
Left BF
Right BF
Worse

Change from Baseline in Static Functional Connectivity to Default Mode Network During the 16-Week Duration Placebo-Controlled Phase

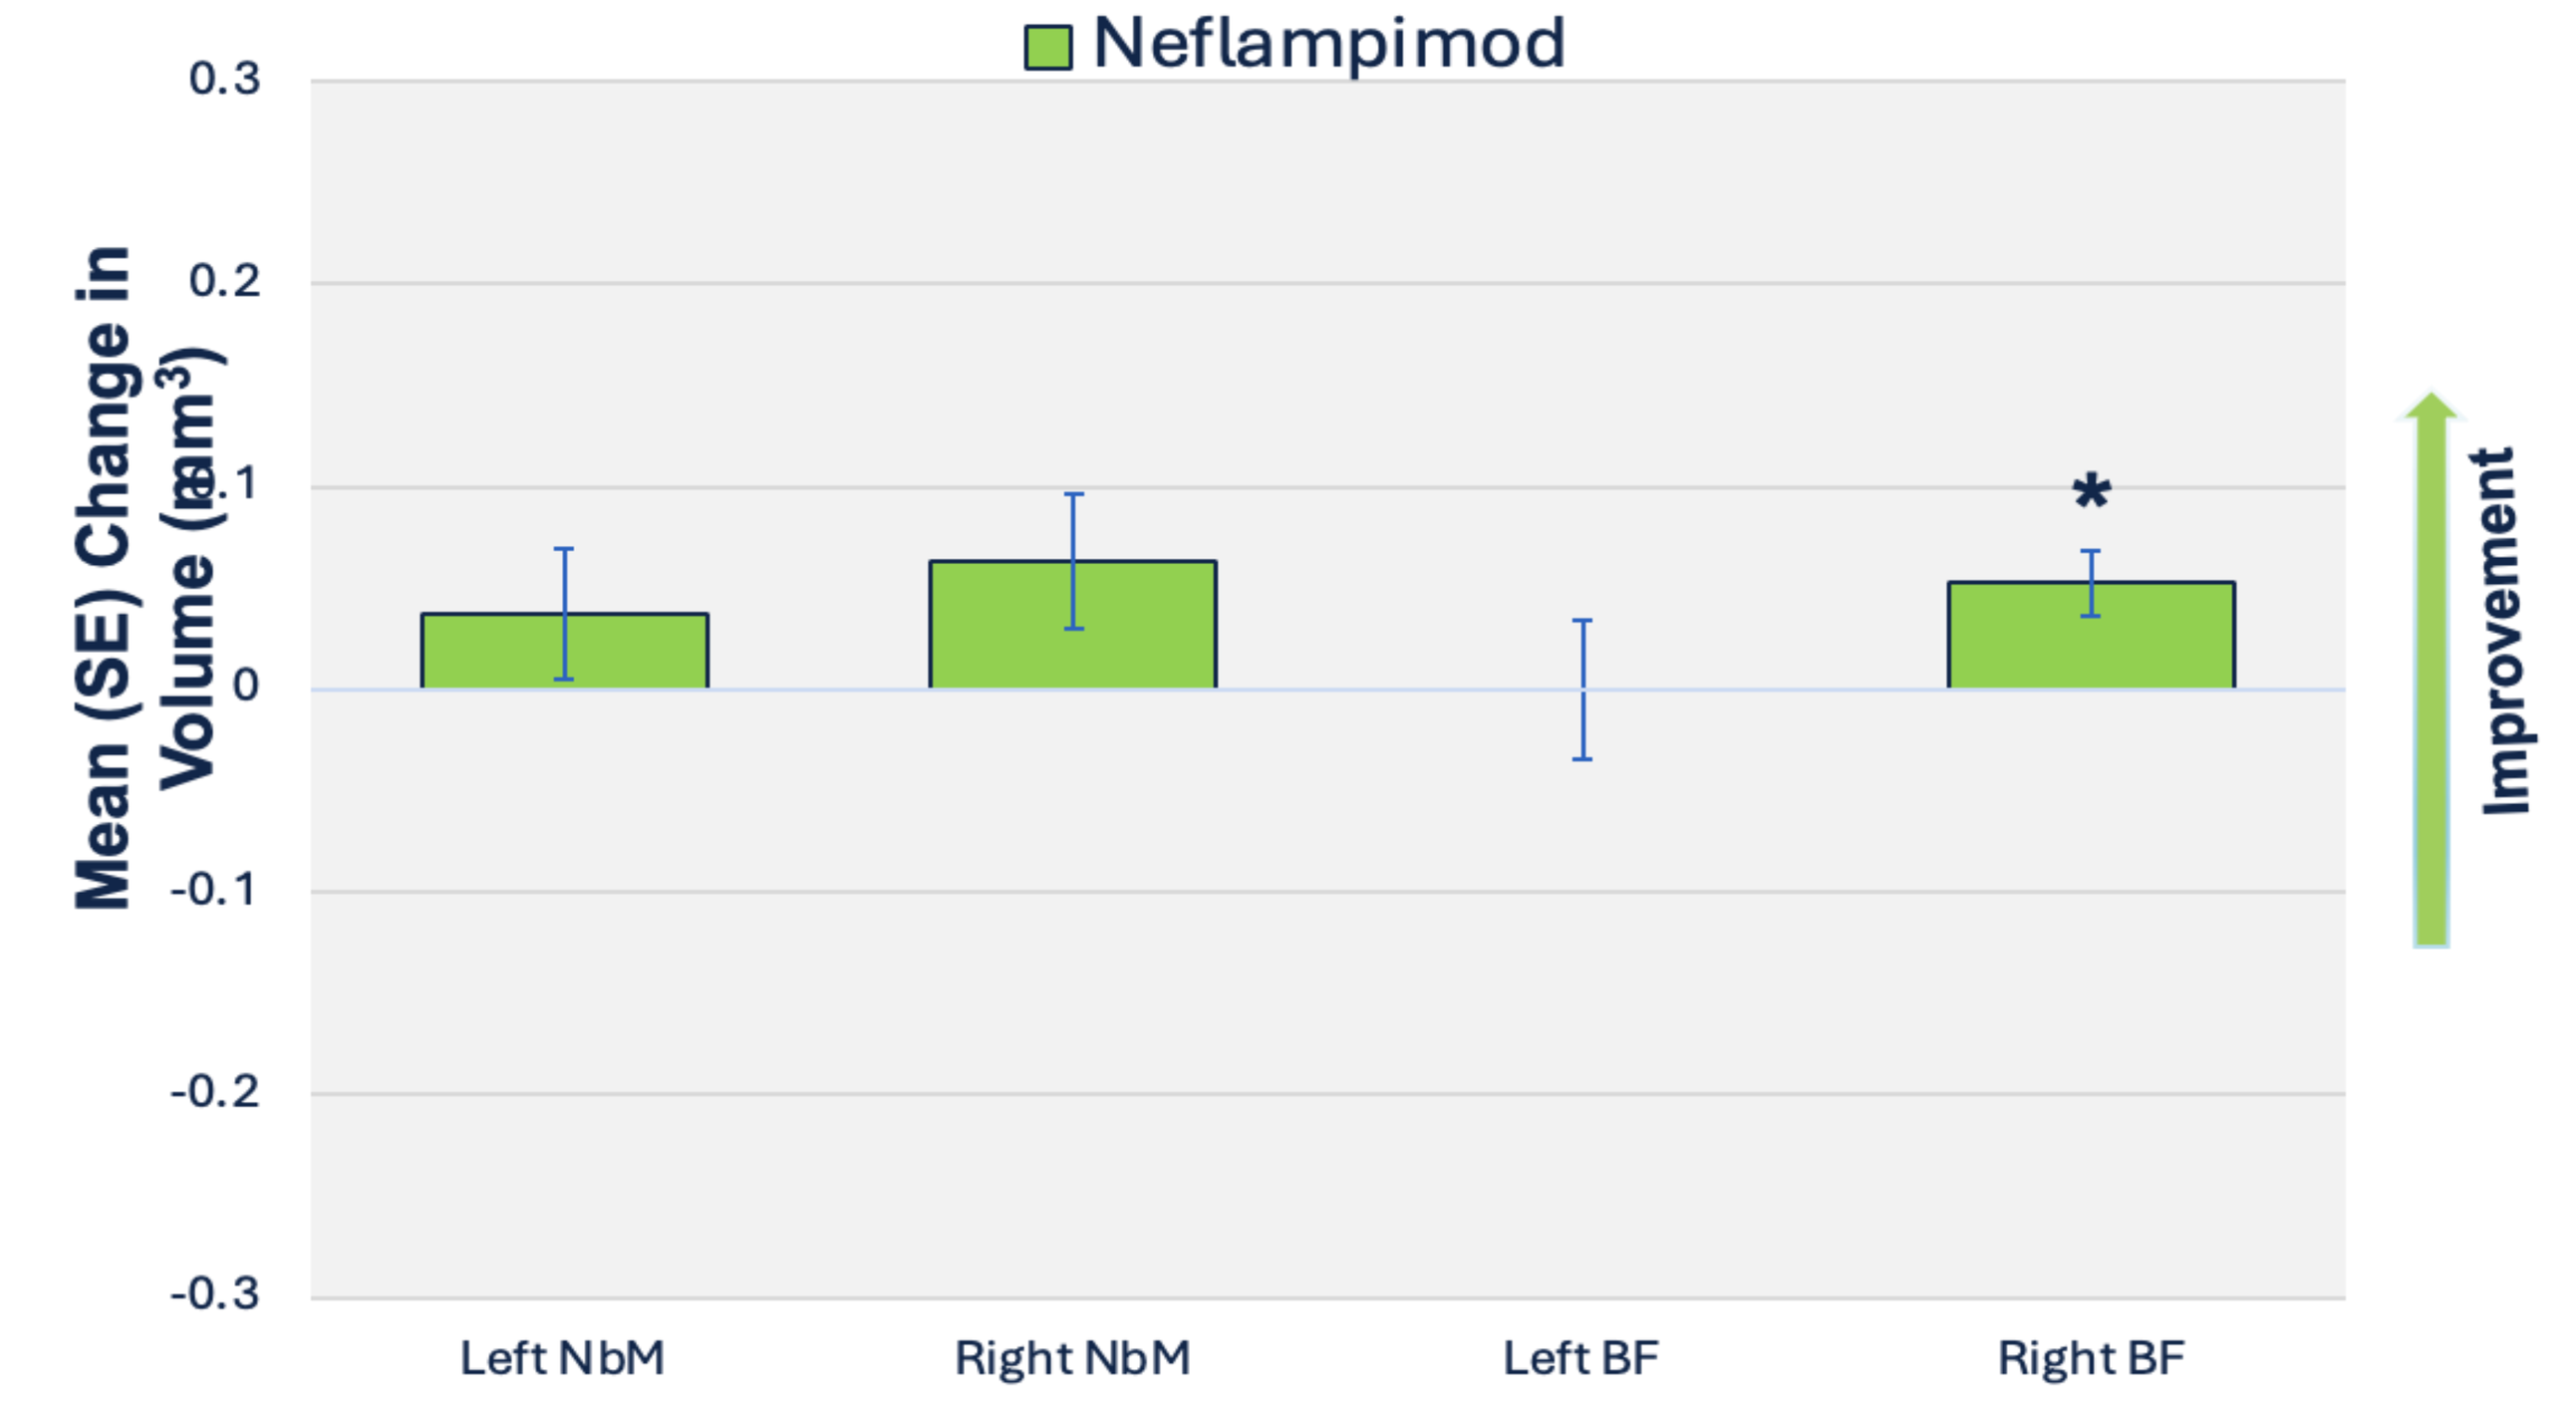
Neflampimod
Mean (SE) Change in Volume (mm³)
0.3
0.2
0.1
0
-0.1
-0.2
-0.3
Left NbM
Right NbM
Left BF
Right BF
*
Improvement

*P=0.019 for change from baseline

NbM – nucleus basalis of Meynert BF – Basal Forebrain

% Change in Right Basal Forebrain Volume Over 16 Weeks

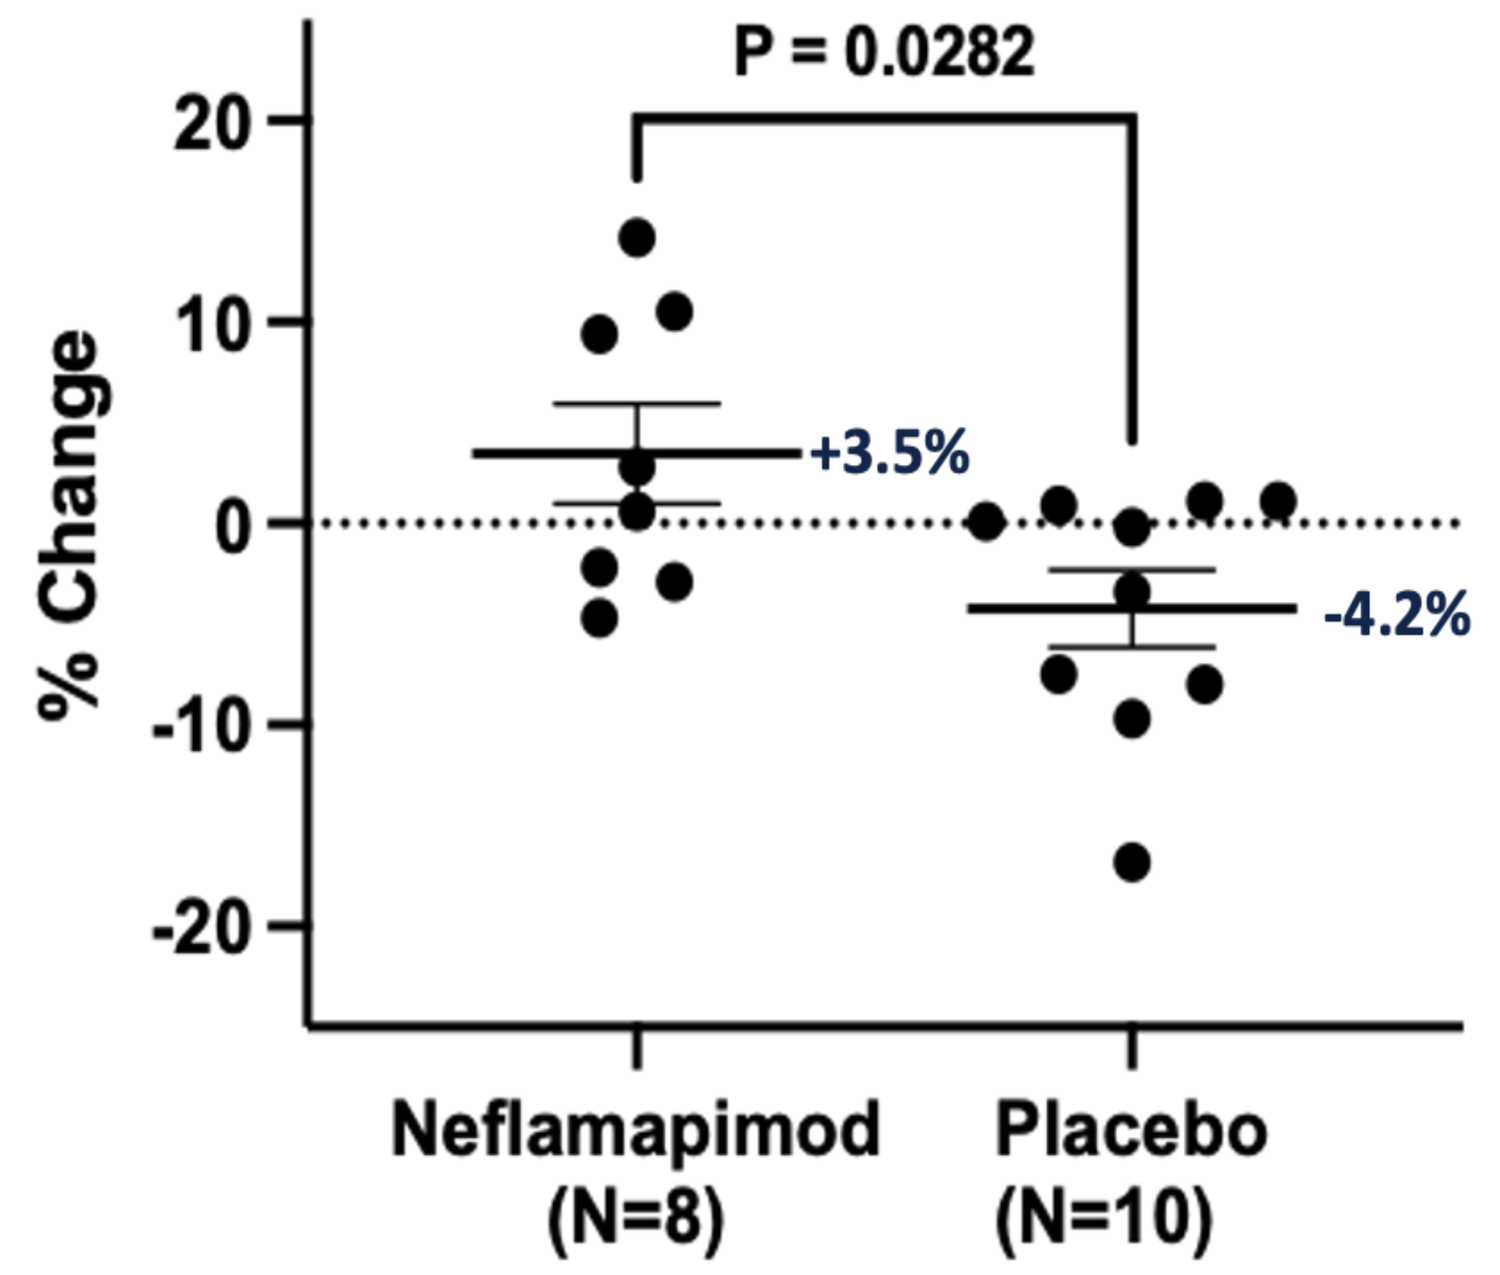
P = 0.0282
% Change
20
10
0
-10
-20
+3.5%
-4.2%
Neflamapimod (N=8)
Placebo (N=10)

Placebo for 16 weeks, followed by 32 weeks Neflamapimod

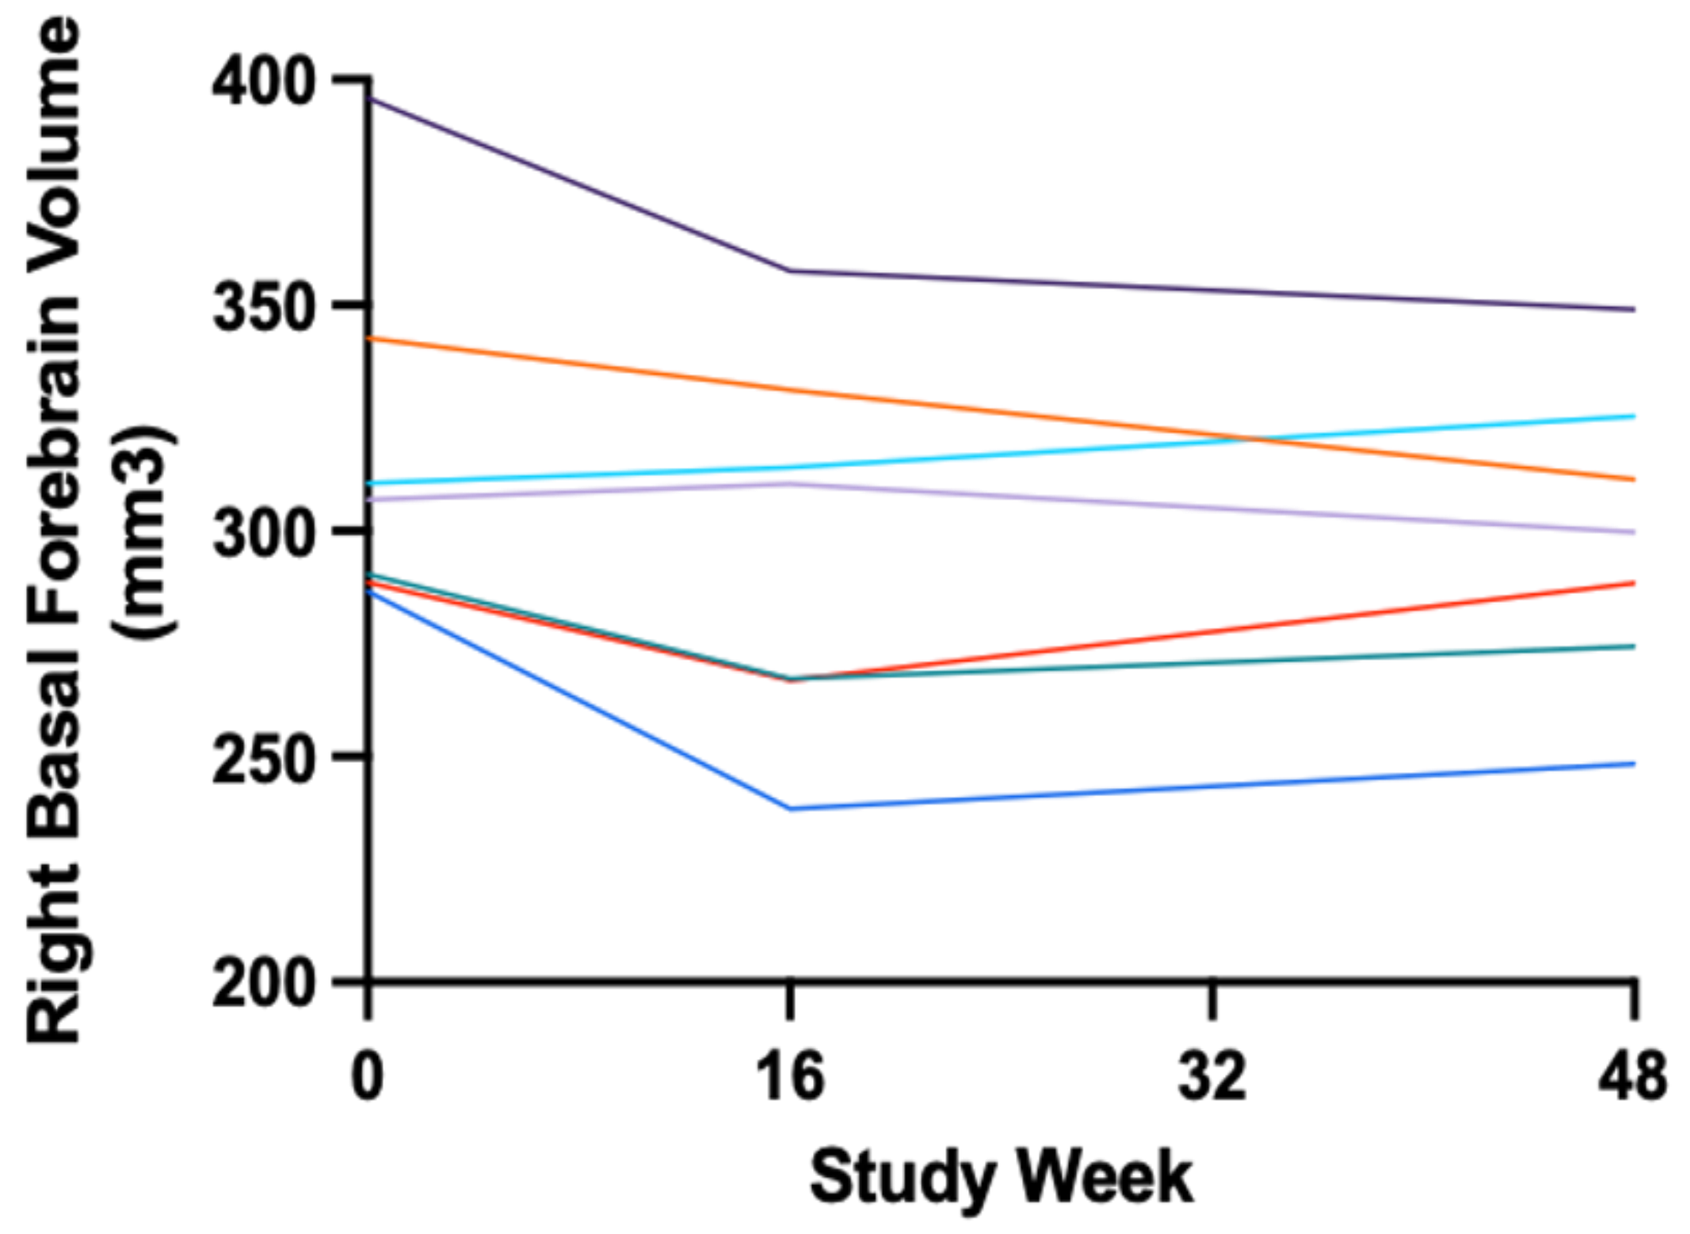
Right Basal Forebrain Volume (mm3)
400
350
300
250
200
0
16
32
48
Study Week

Neflamapimod Over 48 Weeks of Treatment (N=7)

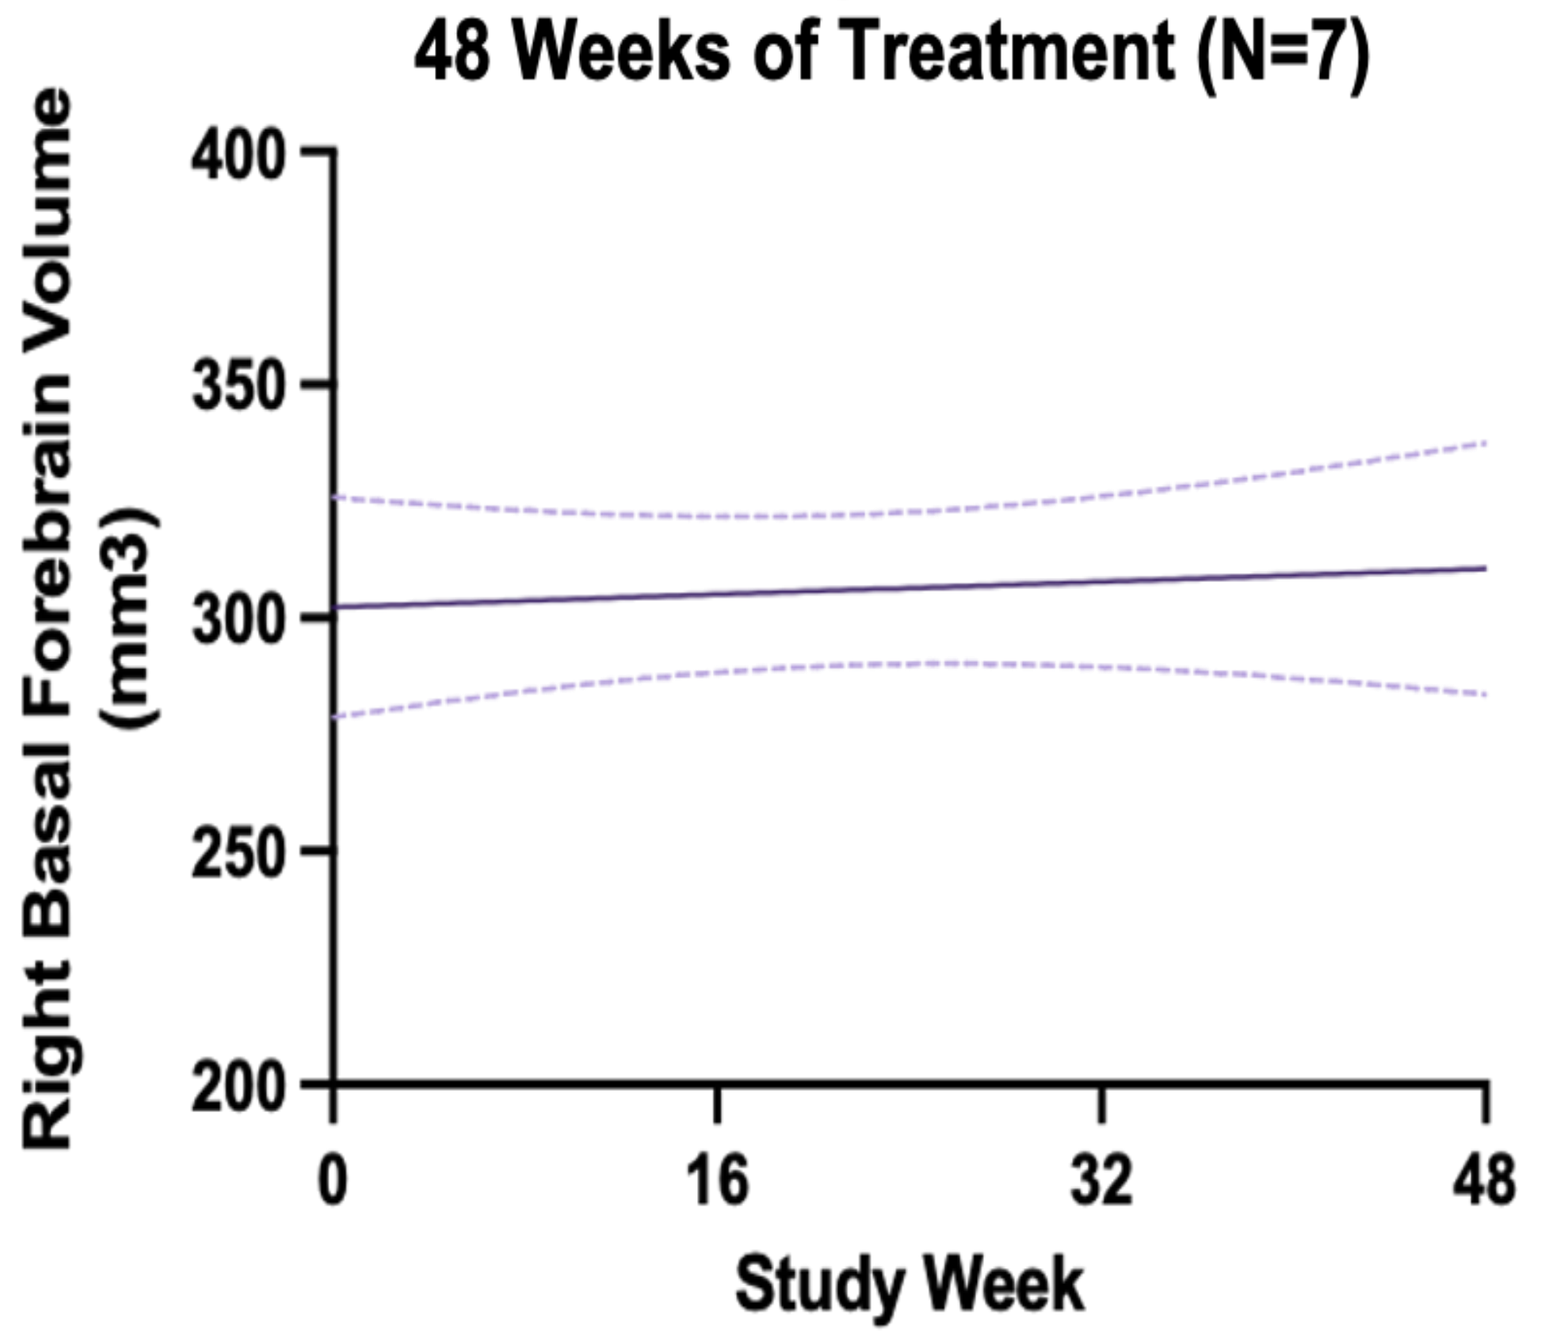
Right Basal Forebrain Volume (mm3)
400
350
300
250
200
0
16
32
48
Study Week

16-week placebo results (N=10) projected out to 48 Weeks

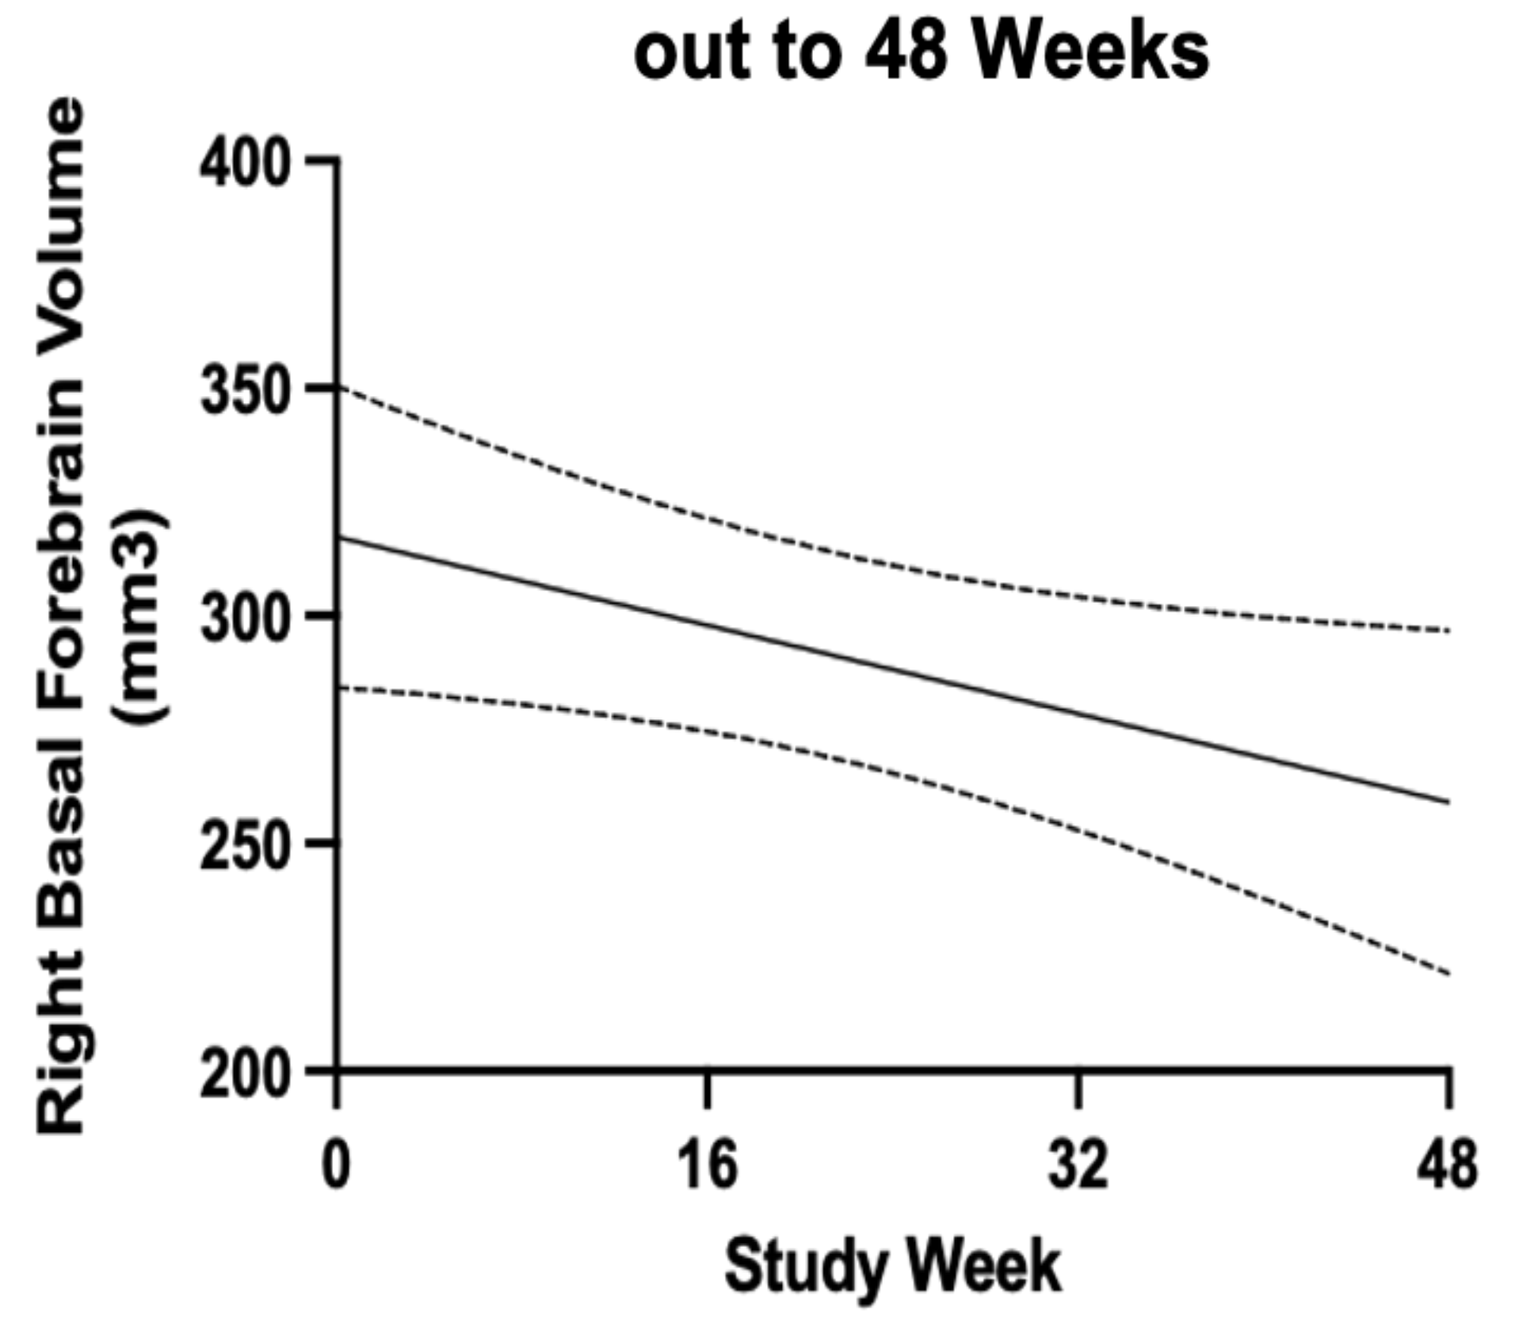
Right Basal Forebrain Volume (mm3)
400
350
300
250
200
0
16
32
48
Study Week

Liner regression lines with 95% confidence intervals are shown

DISCUSSION

- Combined with prior findings in early AD, these results provide evidence that neflamapimod favorably impacts basal forebrain atrophy, consistent with its proposed mechanism of action and preclinical data.
- The observed improvement aligns with animal studies showing that basal forebrain atrophy reflects both neuronal loss and reduced neuronal volume, and with preclinical evidence that neflamapimod reverses cholinergic neuron volume loss.
- The laterality of the treatment effect is consistent with published data that in DLB the left basal forebrain is more severely impacted (Churchill, 2026; Kanek, 2021)
- Right basal forebrain volume remained stable over 48 weeks in participants receiving neflamapimod and stabilized after treatment initiation in prior placebo recipients, suggesting that neflamapimod may slow the progression of basal forebrain atrophy.

ALZHEIMER'S ASSOCIATION | AAIC 26